#8643

Comparable Efficacy and Safety of Taletrectinib for Advanced *ROS1*+ Non-small Cell Lung Cancer Across Pivotal Studies and Between Races and World Regions

Maurice Pérol,[1] Wei Li,[2] Nathan A. Pennell,[3] Geoffrey Liu,[4] Filippo De Braud,[5] Misako Nagasaka,[6] Enriqueta Felip,[7] Anwen Xiong,[2] Yongchang Zhang,[8] Huijie Fan,[9] Xicheng Wang,[10] Feiwu Ran,[11] Xianyu Zhang,[11] Wenfeng Chen,[11] Wei Wang,[11] Lyudmila Bazhenova,[12] Caicun Zhou[2,13]

[1]Department of Medical Oncology, Léon Bérard Cancer Center, Lyon, France; [2]Department of Medical Oncology, Shanghai Pulmonary Hospital and Thoracic Cancer Institute, Tongji University School of Medicine, Shanghai, China; [3]Cleveland Clinic Taussig Cancer Institute, Cleveland, OH, USA; [4]Princess Margaret Cancer Centre, Temerty School of Medicine, University of Toronto, Toronto, Canada; [5]University of Milan, Milan, Italy; [6]University of California Irvine School of Medicine and Chao Family Comprehensive Cancer Center, Orange, CA, USA; [7]Vall d'Hebron University Hospital, Barcelona, Spain; [8]Hunan Cancer Hospital, The Affiliated Cancer Hospital of Xiangya School of Medicine, Central South University, Changsha, China; [9]The First Affiliated Hospital of Zhengzhou University, Zhengzhou, China; [10]The First Affiliated Hospital/School of Clinical Medicine Guangdong Pharmaceutical University, Guangzhou, China; [11]Nuvation Bio, New York, NY, USA; [12]University of California San Diego Moores Cancer Center, San Diego, CA, USA [13]Department of Medical Oncology, East Hospital, Tongji University School of Medicine, Shanghai, China

Supplementary Materials

Copies of this poster and supplementary materials obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the author of this poster.

Background & Methods

- Taletrectinib is an oral, potent, CNS-active, selective, next-generation ROS1 TKI that was evaluated in 2 pivotal *ROS1*+ NSCLC phase 2 trials: the global TRUST-II (NCT04919811) and regional TRUST-I (NCT04395677) studies
- The study designs of TRUST-II[1] and TRUST-I[2] have been previously published
- Here, we compare the efficacy and safety of taletrectinib within and between the pivotal global TRUST-II and regional TRUST-I studies through predefined subgroup analyses

Conclusions

- Taletrectinib demonstrated similar efficacy and safety across the TRUST-II and TRUST-I trials and across subgroups including race, geographic region, and prior chemotherapy status
- TEAEs of special interest, including GI events, occurred early and decreased over time
- Efficacy was unaffected by dose reduction across the two cohorts

Abbreviations

ALT, alanine aminotransferase; AST, aspartate aminotransferase; CI, confidence interval; CNS, central nervous system; cORR, confirmed objective response rate; DCR, disease control rate; DOR, duration of response; ECOG PS, Eastern Cooperative Oncology Group performance status; GI, gastrointestinal; IRC, independent review committee; m, median; mRECIST, modified Response Evaluation Criteria in Solid Tumors; NR, not reached; NSCLC, non-small cell lung cancer; PFS, progression-free survival; QD, once daily; RECIST, Response Evaluation Criteria in Solid Tumors; REP, response evaluable population; ROS1, c-ros oncogene 1; *ROS1+*, *ROS1*-positive; RR, relative risk; TEAE, treatment-emergent adverse event; TKI, tyrosine kinase inhibitor.

References

1. Pérol M, et al. *J Clin Oncol*. 2025;epub April 3, 2025.
2. Li W, et al. *J Clin Oncol*. 2024;42:2660-2670.

Acknowledgments

- We would like to thank all patients who participated in this study, the study investigators, and their staff
- This study was sponsored by Nuvation Bio Inc. Writing and graphical support were provided by David M. Jensen, PhD, of Peloton Advantage, LLC, an OPEN Health company, and funded by Nuvation Bio

Results

Demographics and Baseline Characteristics[a]

TRUST-I[b] (N=170) TRUST-II[c] (N=105)

TKI naive TKI pretreated

	TRUST-I	TRUST-II
Median age	55 y	57 y
Stage IV disease	93.5%	93.3%
Sex, female	57.6%	55.2%
Baseline ECOG PS=1	77.1%	56.2%

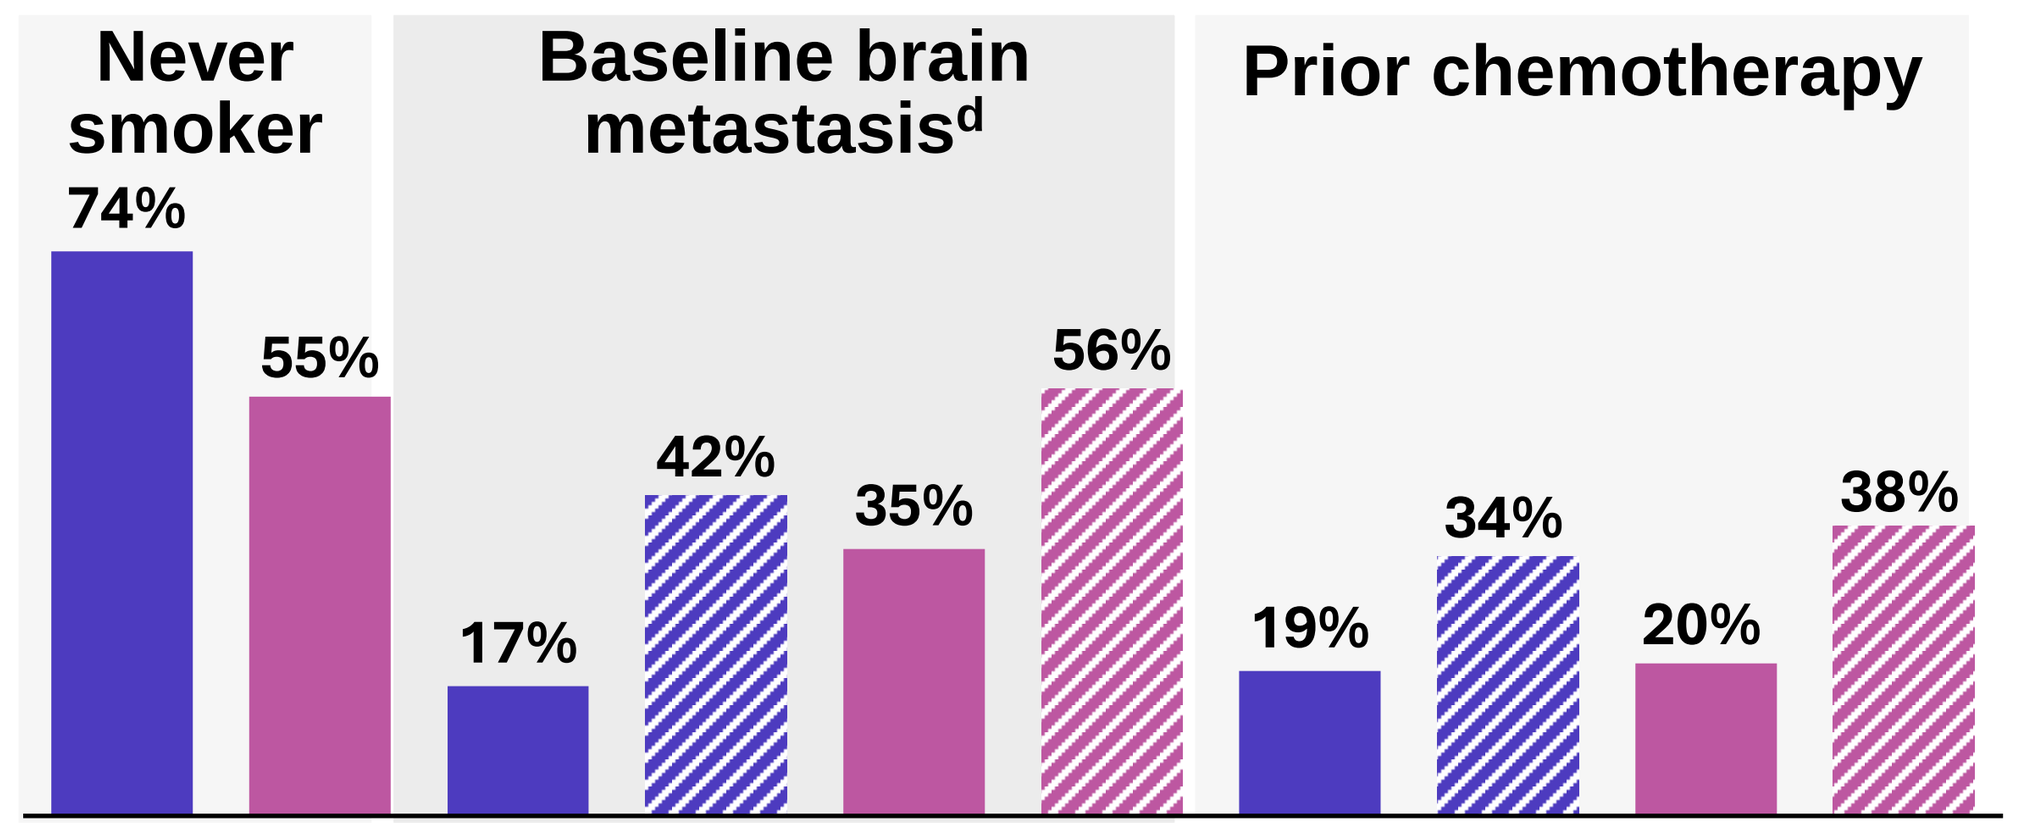
Never smoker
74%
55%
Baseline brain metastasis[d]
17%
42%
35%
56%
Prior chemotherapy
19%
34%
20%
38%

Data cutoff: October 28, 2024

[a]In patients treated with taletrectinib 600 mg QD. [b]TRUST-I was conducted in China. [c]The TRUST-II population included only TKI-naive and TKI-pretreated patients, 47% of whom were geographically Western (North America or Europe) and 53% were Asian (Japan: 24%; Korea: 16%; China: 13%). [d]By IRC per mRECIST v1.1.

TRUST-II and TRUST-I: Efficacy

- Efficacy was assessed in the REP which included patients with ≥1 measurable baseline lesion per RECIST v1.1 who initiated taletrectinib treatment at 600 mg QD across TKI-naive (n=157) and TKI-pretreated (n=113) cohorts in TRUST-II and TRUST-I

TRUST-II and TRUST-I: Comparable Efficacy Across Studies[a]

Endpoint	TRUST-II	TRUST-I	RR (95% CI)
cORR			
TKI naive	46/54 (85.2)	93/103 (90.3)	0.94 (0.83, 1.07)
With prior chemotherapy	9/10 (90.0)	17/20 (85.0)	1.06 (0.80, 1.40)
Without prior chemotherapy	37/44 (84.1)	76/83 (91.6)	0.92 (0.80, 1.06)
TKI pretreated	29/47 (61.7)	34/66 (51.5)	1.20 (0.87, 1.66)
With prior chemotherapy	15/19 (78.9)	10/23 (43.5)	1.82 (1.08, 3.06)
Without prior chemotherapy	14/28 (50.0)	24/43 (55.8)	0.90 (0.57, 1.41)

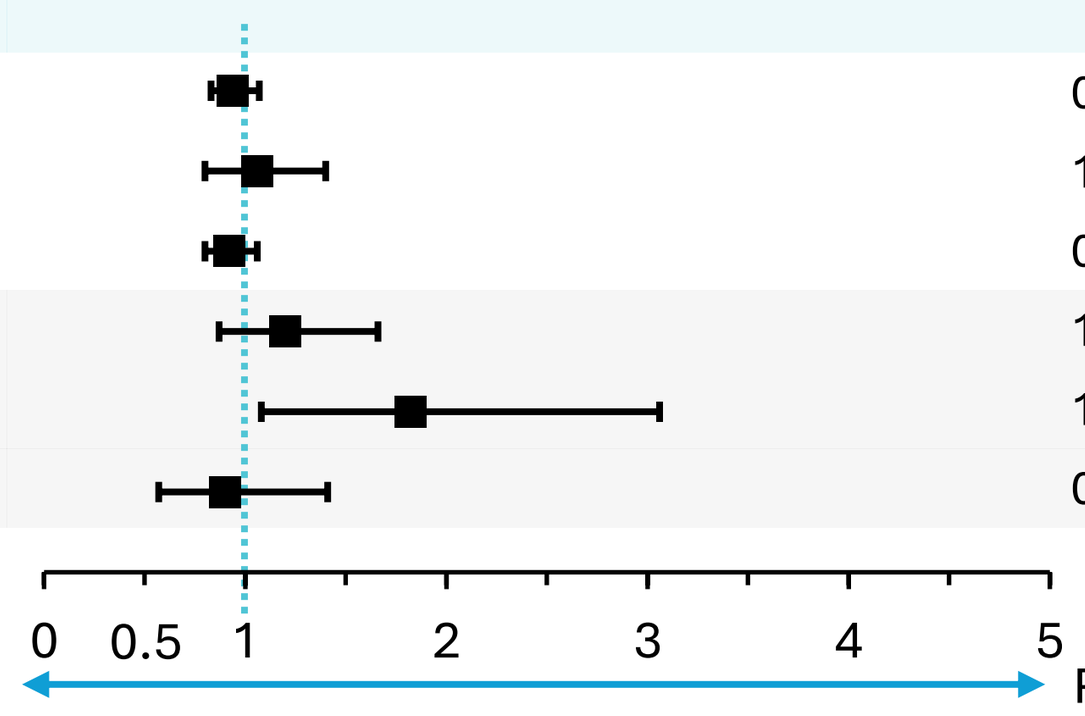
0
0.5
1
2
3
4
5
Favors TRUST-I
Favors TRUST-II

TRUST-II and TRUST-I: Comparable Efficacy Across Race and Region[a]

Race

Endpoint	Non-Asian	Asian	RR (95% CI)
cORR			
TKI naive	16/19 (84.2)	123/138 (89.1)	0.94 (0.77, 1.16)
With prior chemotherapy	6/6 (100)	20/24 (83.3)	1.20 (1.00, 1.44)
Without prior chemotherapy	10/13 (76.9)	103/114 (90.4)	0.85 (0.63, 1.15)
TKI pretreated	16/25 (64.0)	47/88 (53.4)	1.20 (0.84, 1.71)
With prior chemotherapy	8/10 (80.0)	17/32 (53.1)	1.51 (0.96, 2.36)
Without prior chemotherapy	8/15 (53.3)	30/56 (53.6)	1.00 (0.58, 1.70)

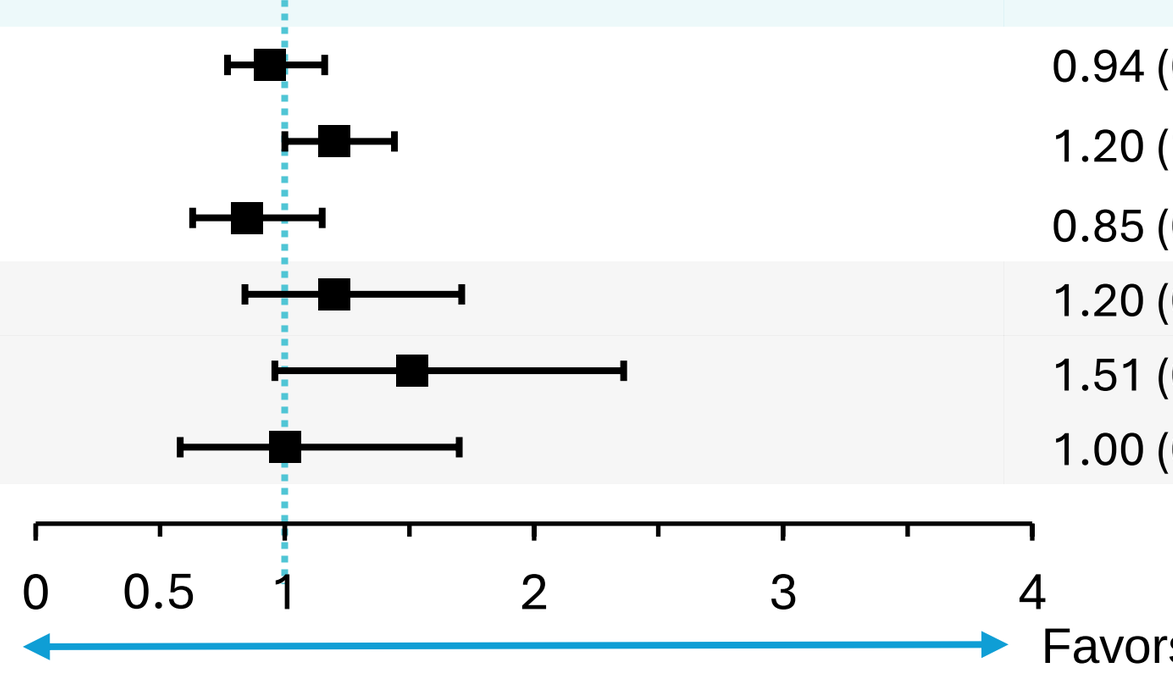
0
0.5
1
2
3
4
Favors Asian
Favors Non-Asian

Region

Endpoint	Western	Non-Western	RR (95% CI)
cORR			
TKI naive	17/21 (81.0)	122/136 (89.7)	0.90 (0.73, 1.12)
With prior chemotherapy	6/6 (100)	20/24 (83.3)	1.20 (1.00, 1.44)
Without prior chemotherapy	11/15 (73.3)	102/112 (91.1)	0.81 (0.59, 1.10)
TKI pretreated	17/26 (65.4)	46/87 (52.9)	1.24 (0.88, 1.74)
With prior chemotherapy	8/10 (80.0)	17/32 (53.1)	1.51 (0.96, 2.36)
Without prior chemotherapy	9/16 (56.3)	29/55 (52.7)	1.07 (0.65, 1.76)

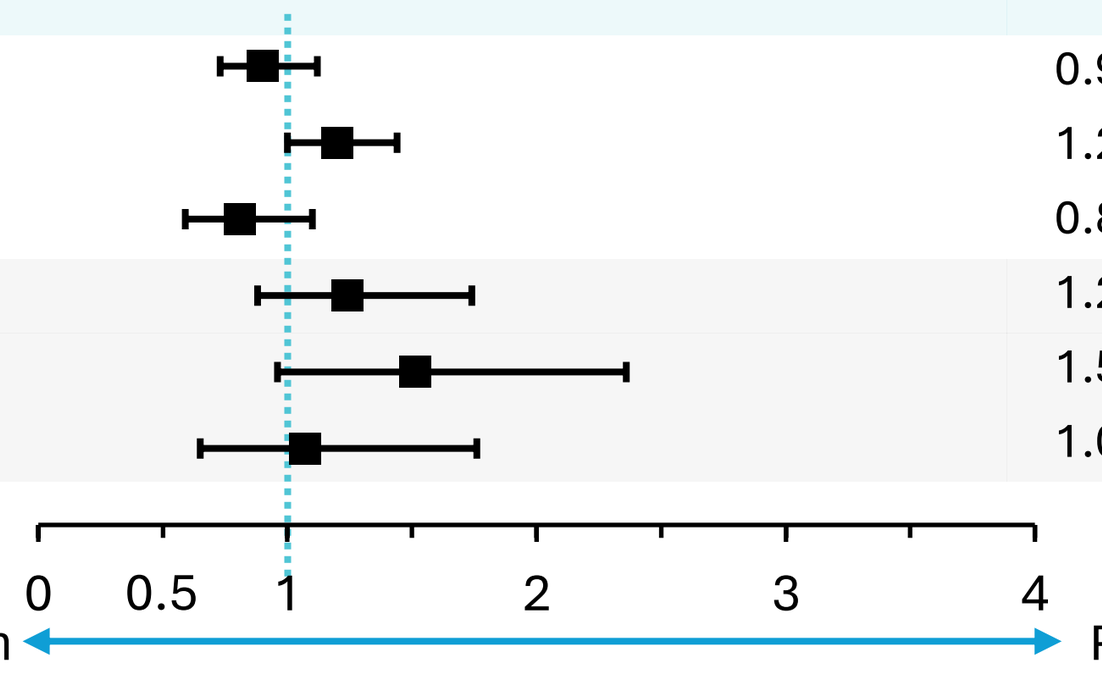
0
0.5
1
2
3
4
Favors Non-Western
Favors Western

[a]An RR of 1 indicates there is no difference between groups.

- Comparable efficacy was observed across race and region within TRUST-II (**Supplement**)

TRUST-II and TRUST-I: Safety

TRUST-II and TRUST-I: Comparable Safety Across Studies[a,b]

- Safety was assessed in all patients who received ≥1 dose of taletrectinib in TRUST-II (n=171)[1] and TRUST-I (n=173)[2]

Endpoint	TRUST-II	TRUST-I	RR (95% CI)
Grade ≥3 TEAE	90/171 (52.6)	94/173 (54.3)	0.97 (0.80, 1.18)
Grade ≥3 ALT/AST elevation	31/171 (18.1)	35/173 (20.2)	0.90 (0.58, 1.38)
Grade ≥3 GI disorders	7/171 (4.1)	10/173 (5.8)	0.71 (0.28, 1.82)

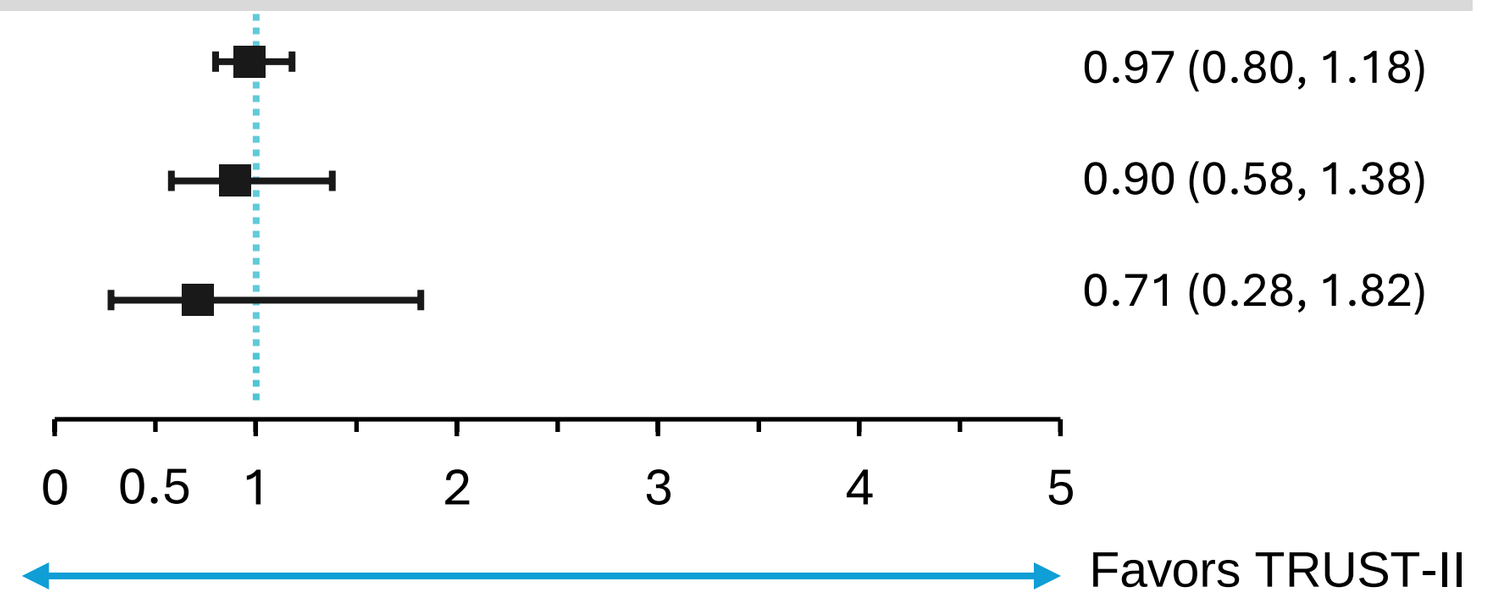
0
0.5
1
2
3
4
5
Favors TRUST-I
Favors TRUST-II

[a]An RR of 1 indicates there is no difference between groups.
[b]The TRUST-II population included patients across cohorts 1 to 5 treated with taletrectinib 600 mg QD.[1]

TRUST-II[a] and TRUST-I: Comparable Safety Across Race and Region[a]

Race

Endpoint	Non-Asian	Asian	RR (95% CI)
Grade ≥3 TEAE	50/91 (54.9)	134/253 (53.0)	1.04 (0.83, 1.29)
Grade ≥3 ALT/AST elevation	19/91 (20.9)	47/253 (18.6)	1.12 (0.70, 1.81)
Grade ≥3 GI disorders	5/91 (5.5)	12/253 (4.7)	1.16 (0.42, 3.20)

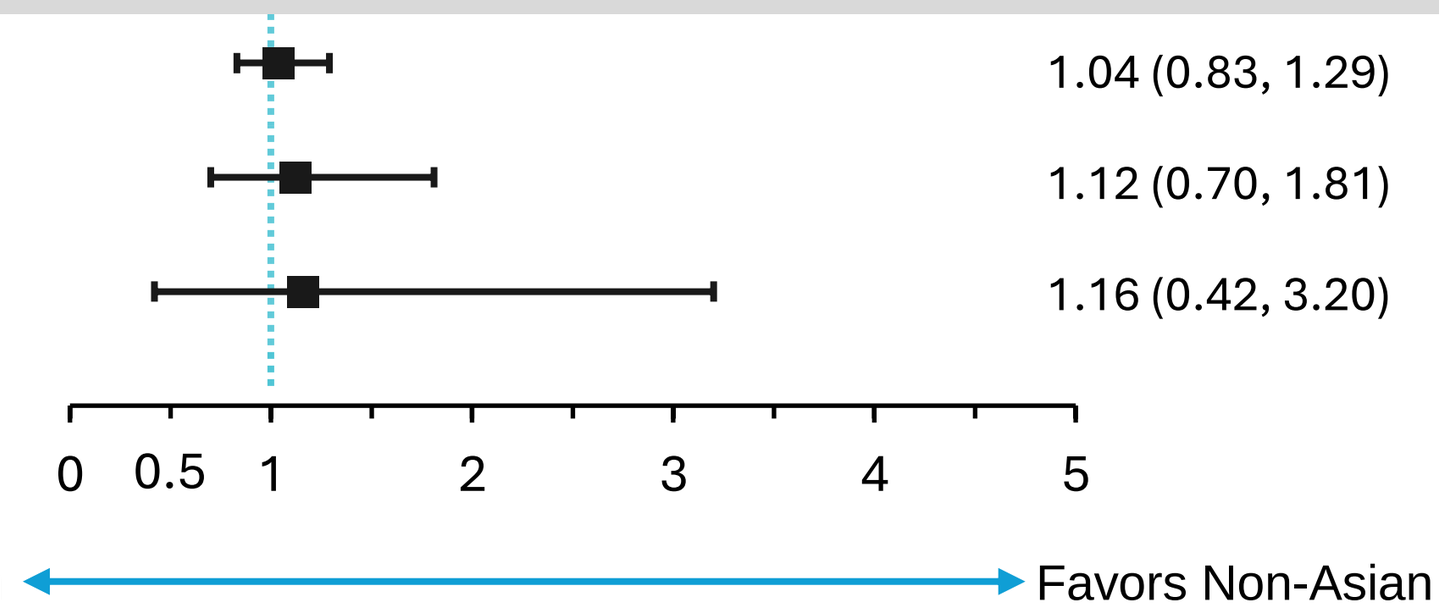
0
0.5
1
2
3
4
5
Favors Asian
Favors Non-Asian

[a]An RR of 1 indicates there is no difference between groups.

Region

Endpoint	Western	Non-Western	RR (95% CI)
Grade ≥3 TEAE	54/97 (55.7)	130/247 (52.6)	1.06 (0.85, 1.31)
Grade ≥3 ALT/AST elevation	20/97 (20.6)	46/247 (18.6)	1.11 (0.69, 1.77)
Grade ≥3 GI disorders	5/97 (5.2)	12/247 (4.9)	1.06 (0.38, 2.93)

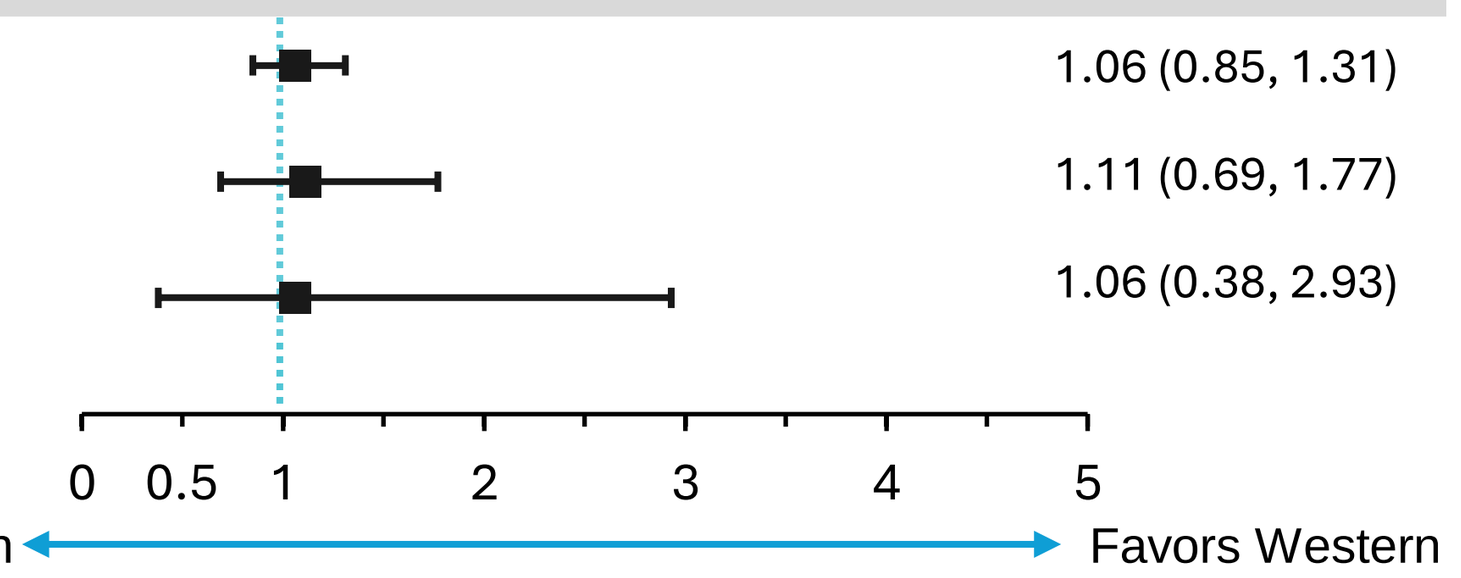
0
0.5
1
2
3
4
5
Favors Non-Western
Favors Western

[a]An RR of 1 indicates there is no difference between groups.

TRUST-II and TRUST-I: TEAEs of Special Interest

- Due to comparable overall safety across TRUST-II and TRUST-I, a detailed safety characterization of TEAEs of special interest was conducted in a combined population from both studies (Table below and **Supplement**)

TRUST-II[a] and TRUST-I: GI Events Across Studies

	TRUST-II (n=171)		TRUST-I (n=173)	
GI Events	All grades, n (%)	Grades ≥3, n (%)	All grades, n (%)	Grades ≥3, n (%)
Diarrhea	99 (57.9)	1 (0.6)	121 (69.9)	6 (3.5)
Vomiting	59 (34.5)	2 (1.2)	94 (54.3)	2 (1.2)
Nausea	89 (52.0)	3 (1.8)	74 (42.8)	2 (1.2)

[a]The TRUST-II population included patients across cohorts 1 to 5 treated with taletrectinib 600 mg QD.[1]

Most GI Events Occurred Early in Treatment and Decreased Over Time

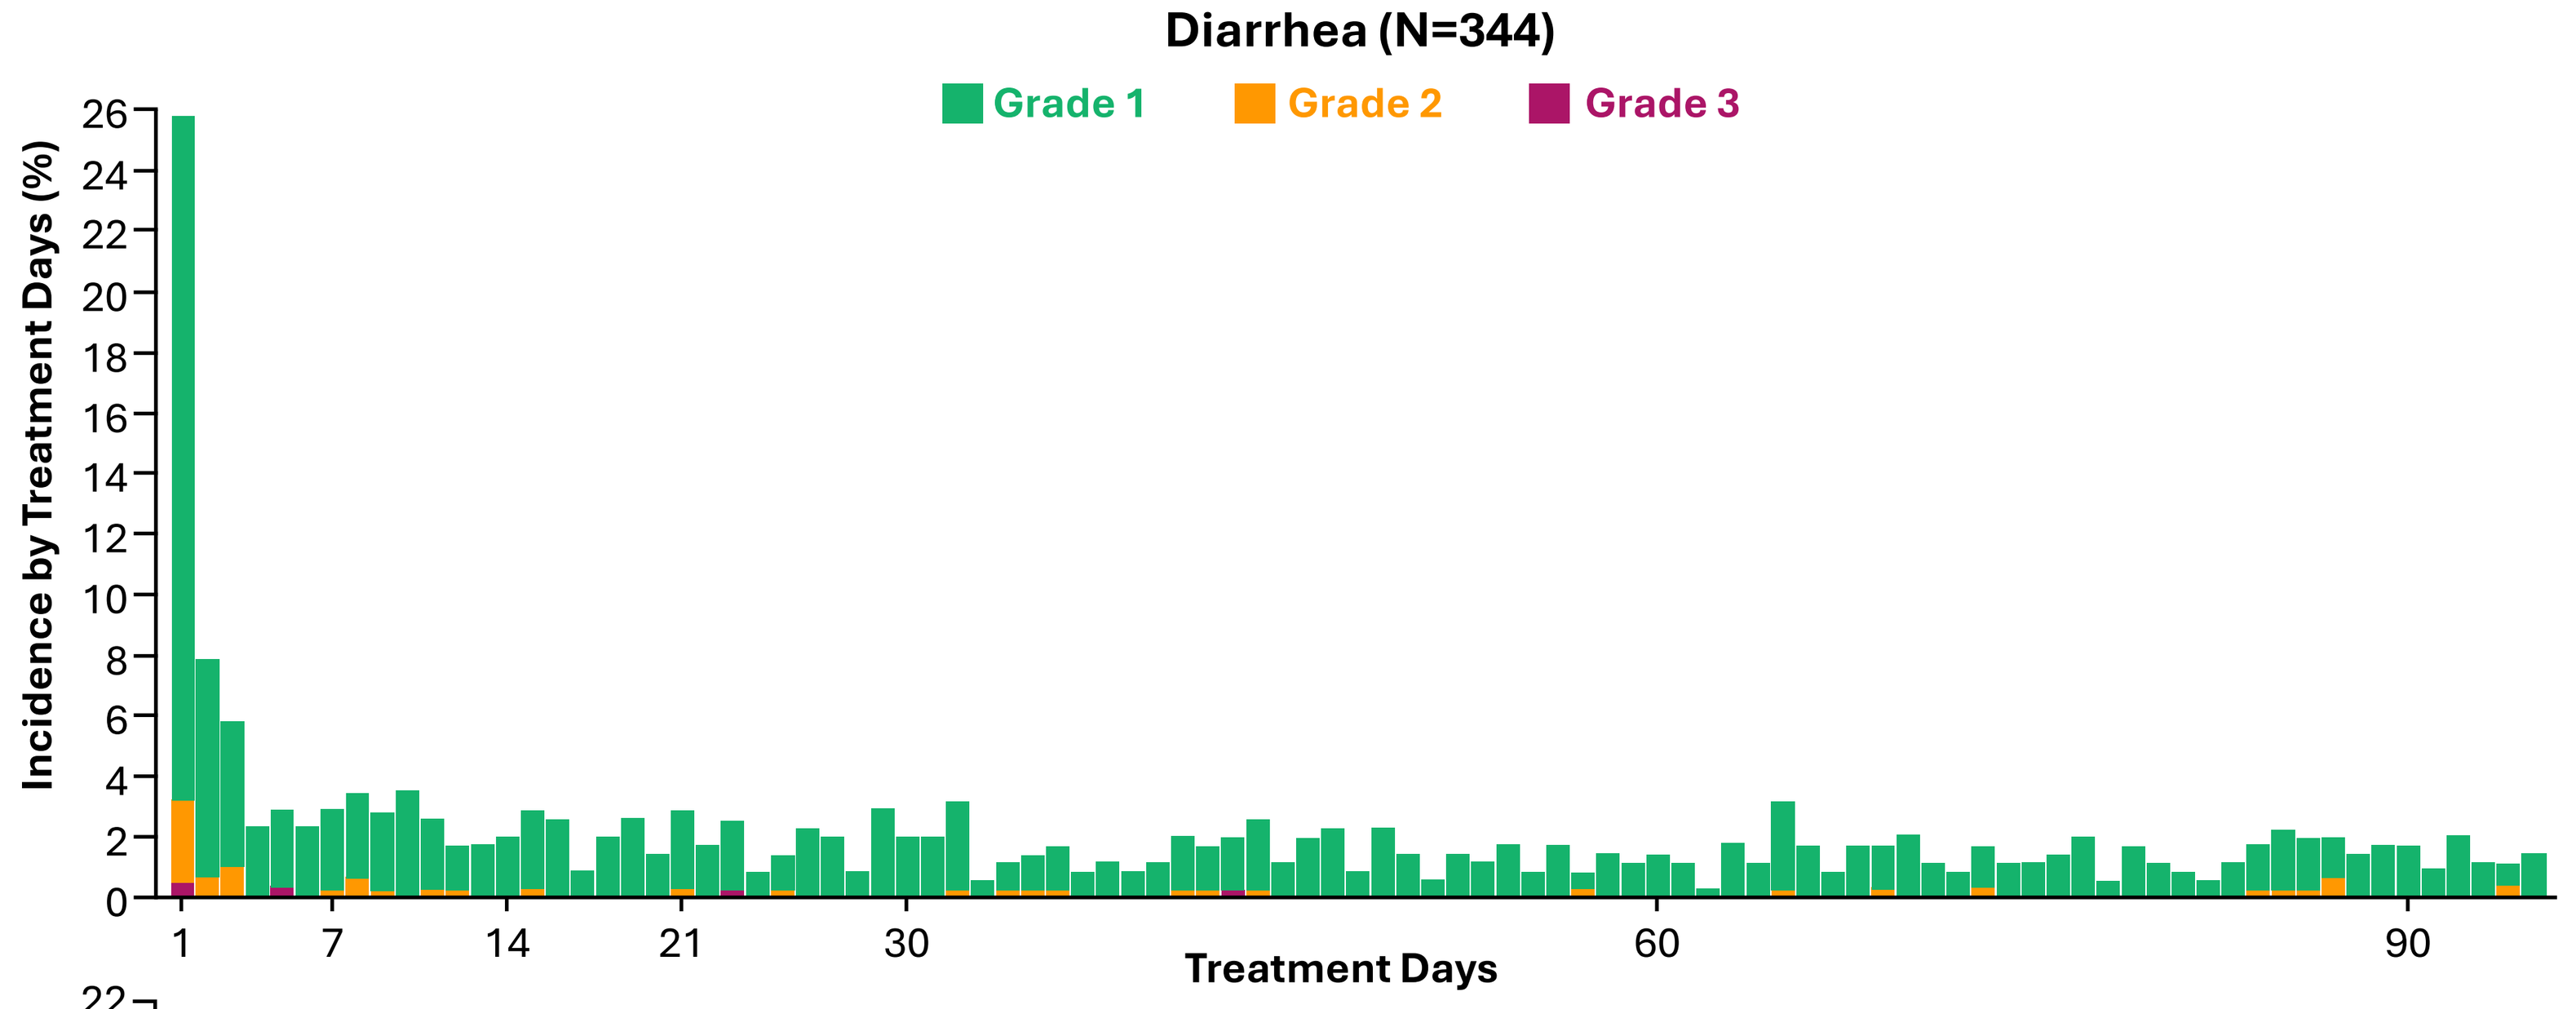
Diarrhea (N=344)
Grade 1
Grade 2
Grade 3
Incidence by Treatment Days (%)
Treatment Days

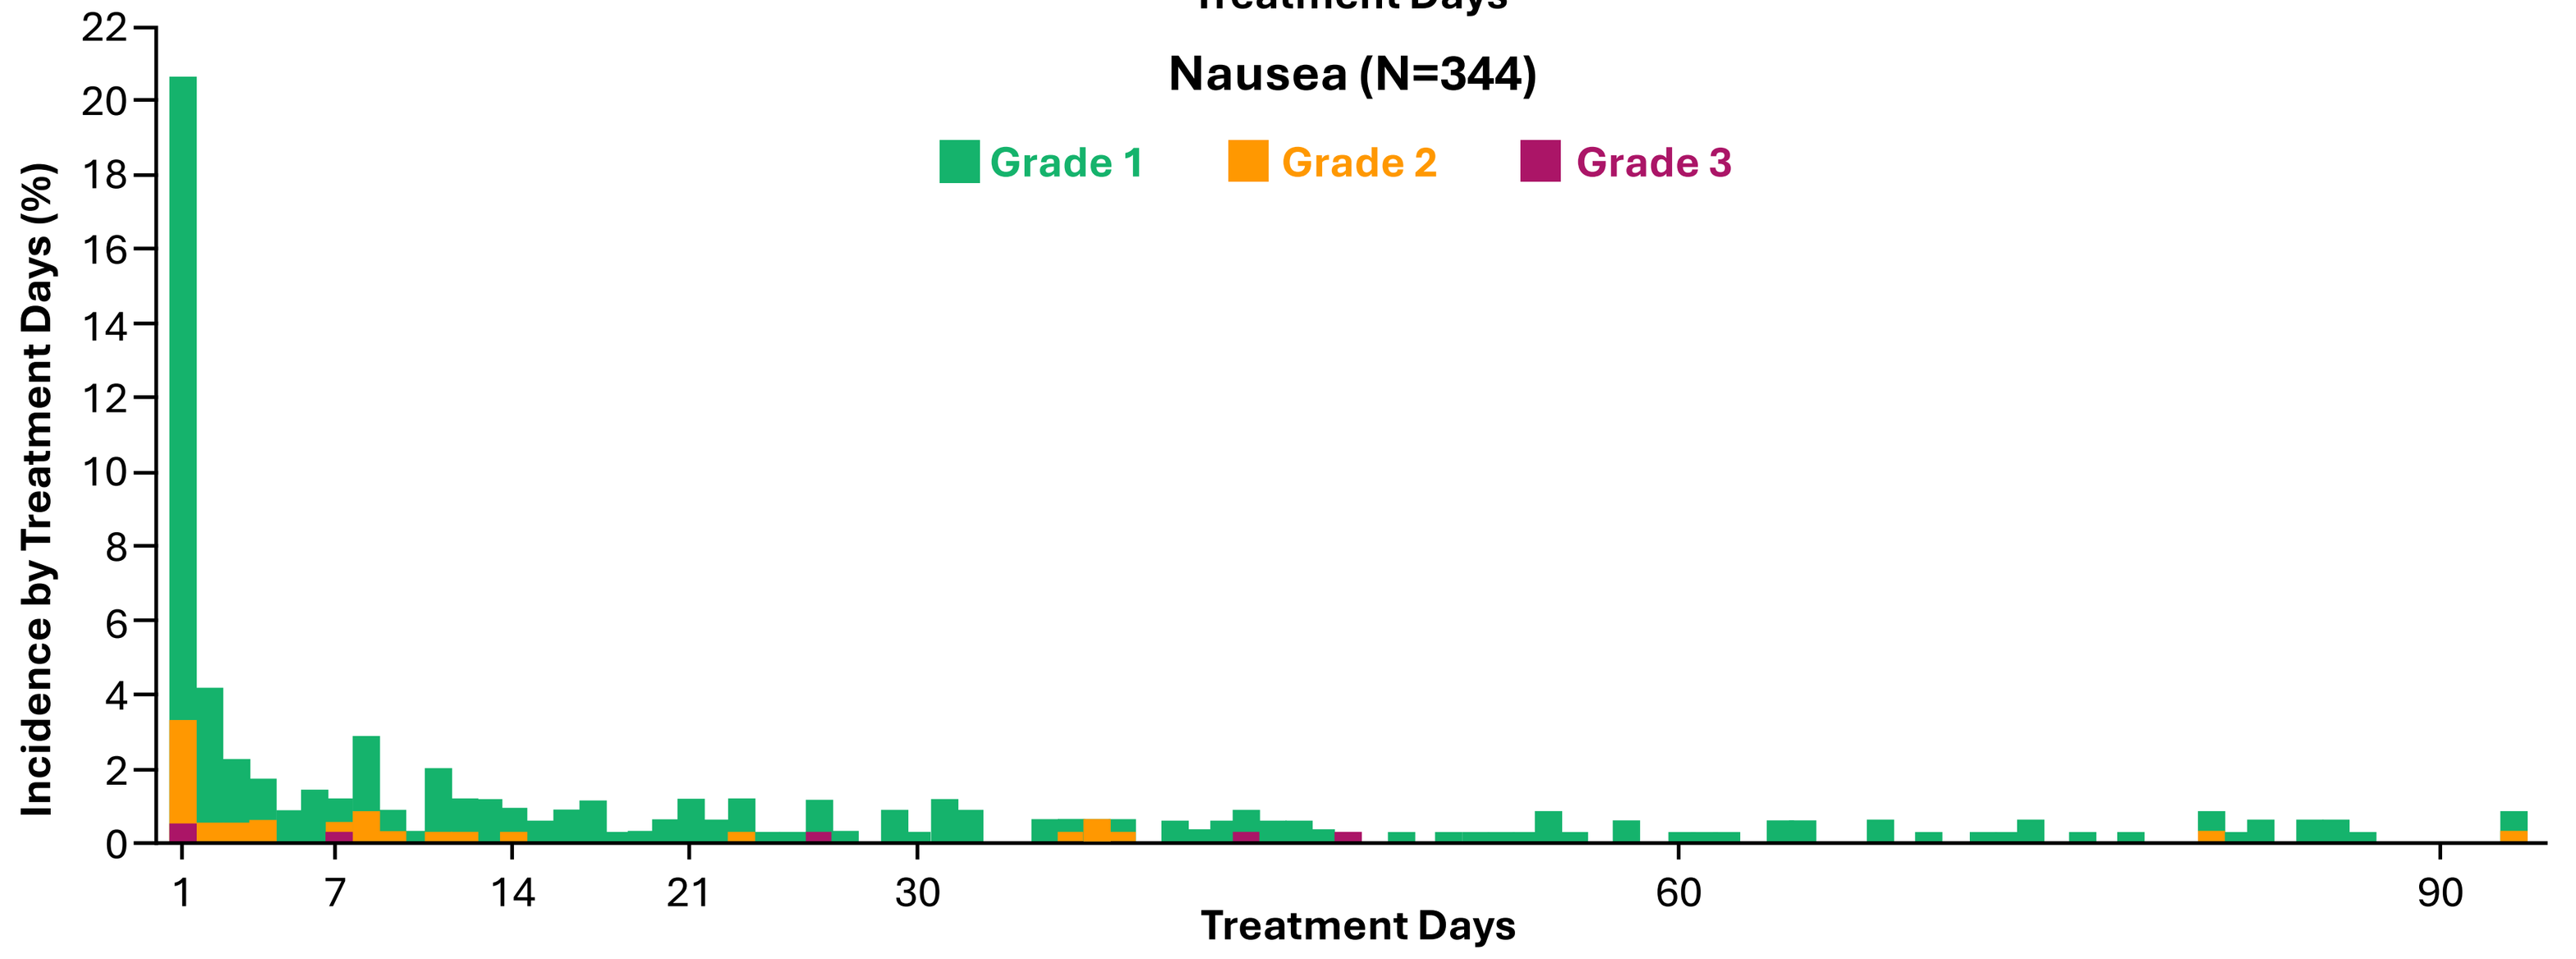
Nausea (N=344)
Grade 1
Grade 2
Grade 3
Incidence by Treatment Days (%)
Treatment Days

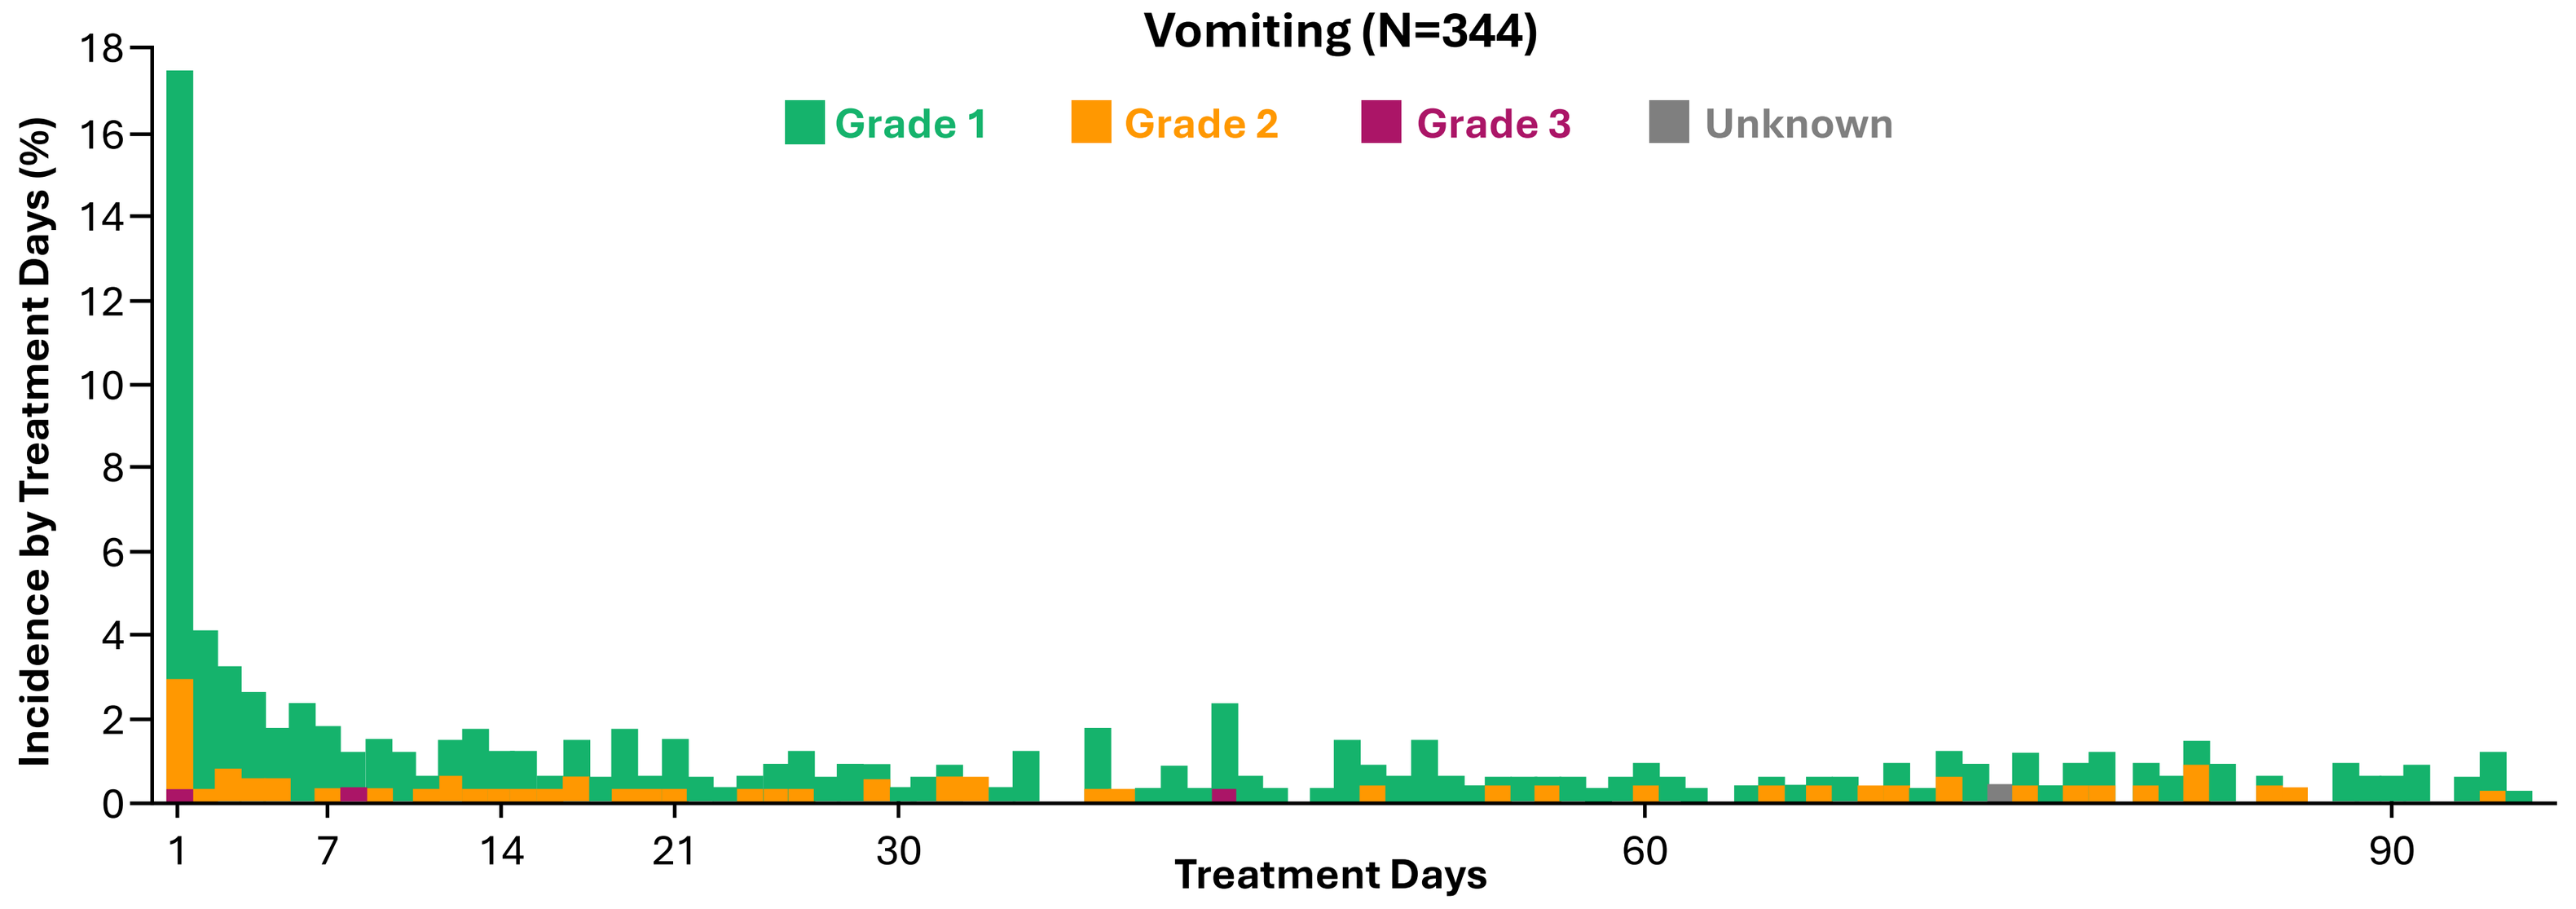
Vomiting (N=344)
Grade 1
Grade 2
Grade 3
Unknown
Incidence by Treatment Days (%)
Treatment Days

Dose Reduction[a] Does Not Compromise Efficacy

- DOR and PFS across the efficacy population were comparable regardless of dose reduction status (**Supplement**)

	TKI-naive (n=157)		TKI-pretreated (n=113)	
	With Dose Reduction	Without Dose Reduction	With Dose Reduction	Without Dose Reduction
Median follow-up time, months	23.9	27.1	22.1	27.3
N	46	111	28	85
cORR,% (95 CI%)	95.7 (85.2, 99.5)	85.6 (77.6, 91.5)	71.4 (51.3, 86.8)	50.6 (39.5, 61.6)
DCR, % (95% CI)	97.8 (88.5, 99.9)	93.7 (87.4, 97.4)	92.9 (76.5, 99.1)	85.9 (76.6, 92.5)
Median DOR, months [95% CI] (Range)	38.6 [14.7, NR] (1.1–46.9)	NR	12.5 [8.9, NR] (4.1–31.8)	16.6 [10.4, 27.3] (1.4–38.7)

[a]Following grade ≥3 increased ALT/AST, patients in TRUST-I could resume taletrectinib at their original dose or a reduced dose while patients in TRUST-II were required to resume taletrectinib at a reduced dose

American Society of Clinical Oncology | May 30–June 3, 2025 | Chicago, IL, USA

TRUST-II: Comparable Efficacy Across Race, Region, and Prior Chemotherapy in Patients with Advanced *ROS1*+ NSCLC Treated with Taletrectinib 600 mg QD

TKI Naive

Subgroup	N	Patients with a Response	cORR (95% CI)	cORR (95% CI)
Overall	54	46		85.2 (72.88, 93.38)
Region				
Western	21	17		81.0 (58.09, 94.55)
Non-Western	33	29		87.9 (71.80, 96.60)
Race				
Non-Asian	19	16		84.2 (60.42, 96.62)
Asian	35	30		85.7 (69.74, 95.19)
Prior chemotherapy				
Yes	10	9		90.0 (55.50, 99.75)
No	44	37		84.1 (69.93, 93.36)

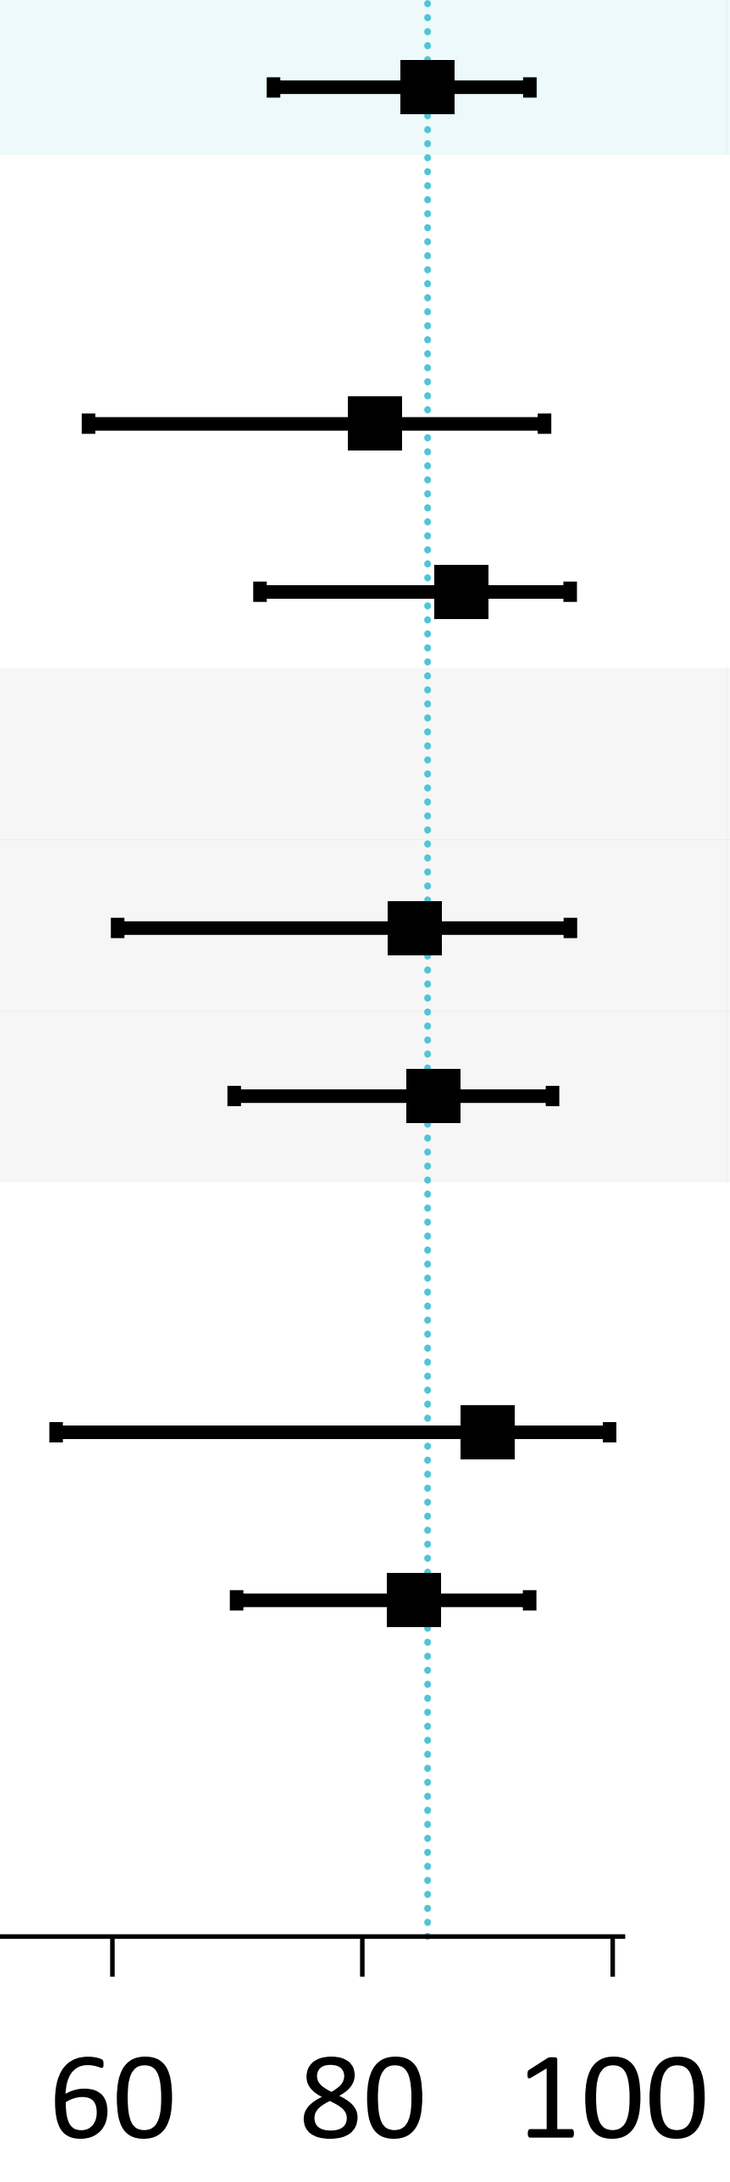
0
20
40
60
80
100

TKI Pretreated

Subgroup	N	Patients with a Response	cORR (95% CI)	cORR (95% CI)
Overall	47	29		61.7 (46.38, 75.49)
Region				
Western	26	17		65.4 (44.33, 82.79)
Non-Western	21	12		57.1 (34.02, 78.18)
Race				
Non-Asian	25	16		64.0 (42.52, 82.03)
Asian	22	13		59.1 (36.35, 79.29)
Prior chemotherapy				
Yes	19	15		78.9 (54.43, 93.95)
No	28	14		50.0 (30.65, 69.35)

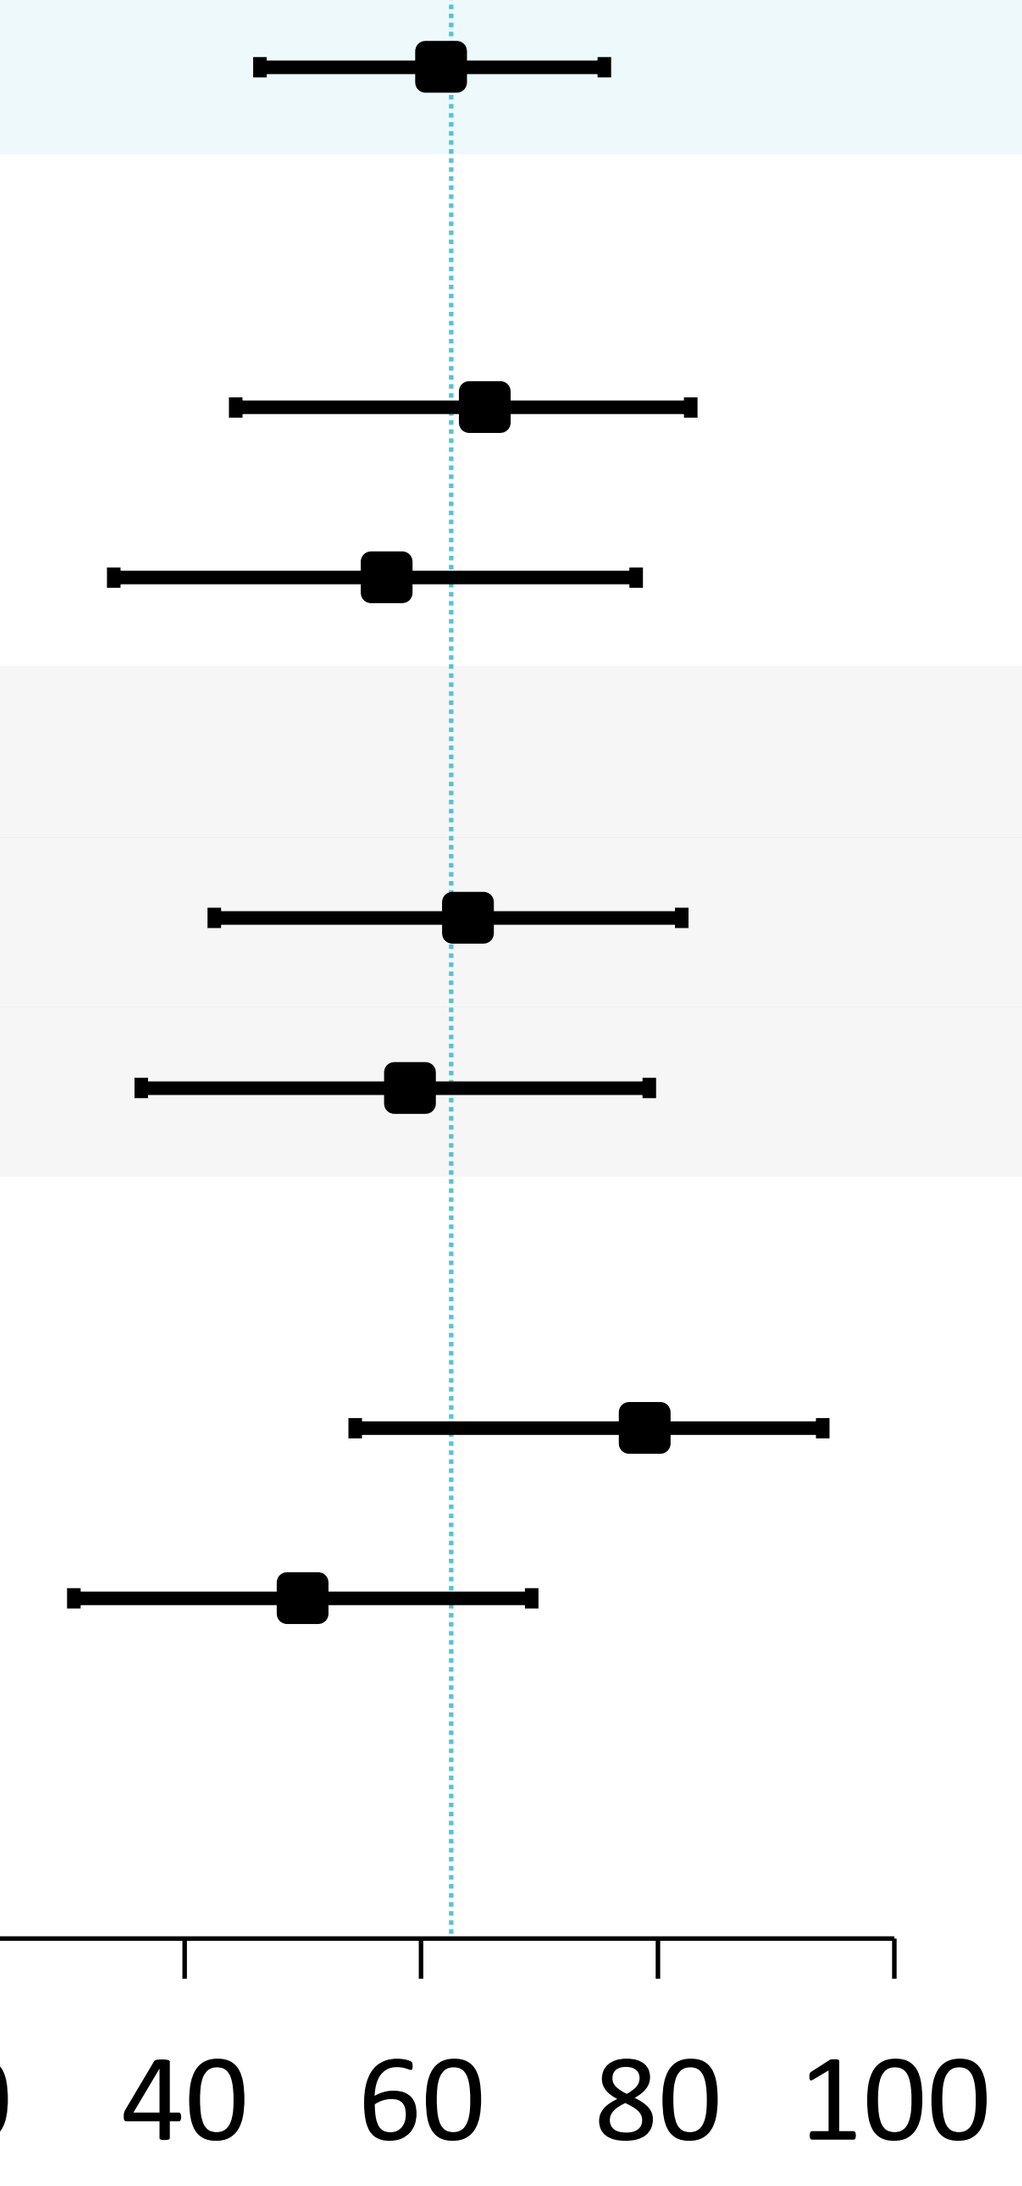
0
20
40
60
80
100

Data cutoff: October 28, 2024.
Dashed vertical line represents the overall cORR for TKI-naive patients.
CI, confidence interval; cORR, confirmed objective response rate; NSCLC, non-small cell lung cancer; QD, once daily; TKI, tyrosine kinase inhibitor.

TRUST-II and TRUST-I[a]: DOR and PFS Across the Efficacy Population Were Similar Regardless of Dose Reduction Status

TKI Naive

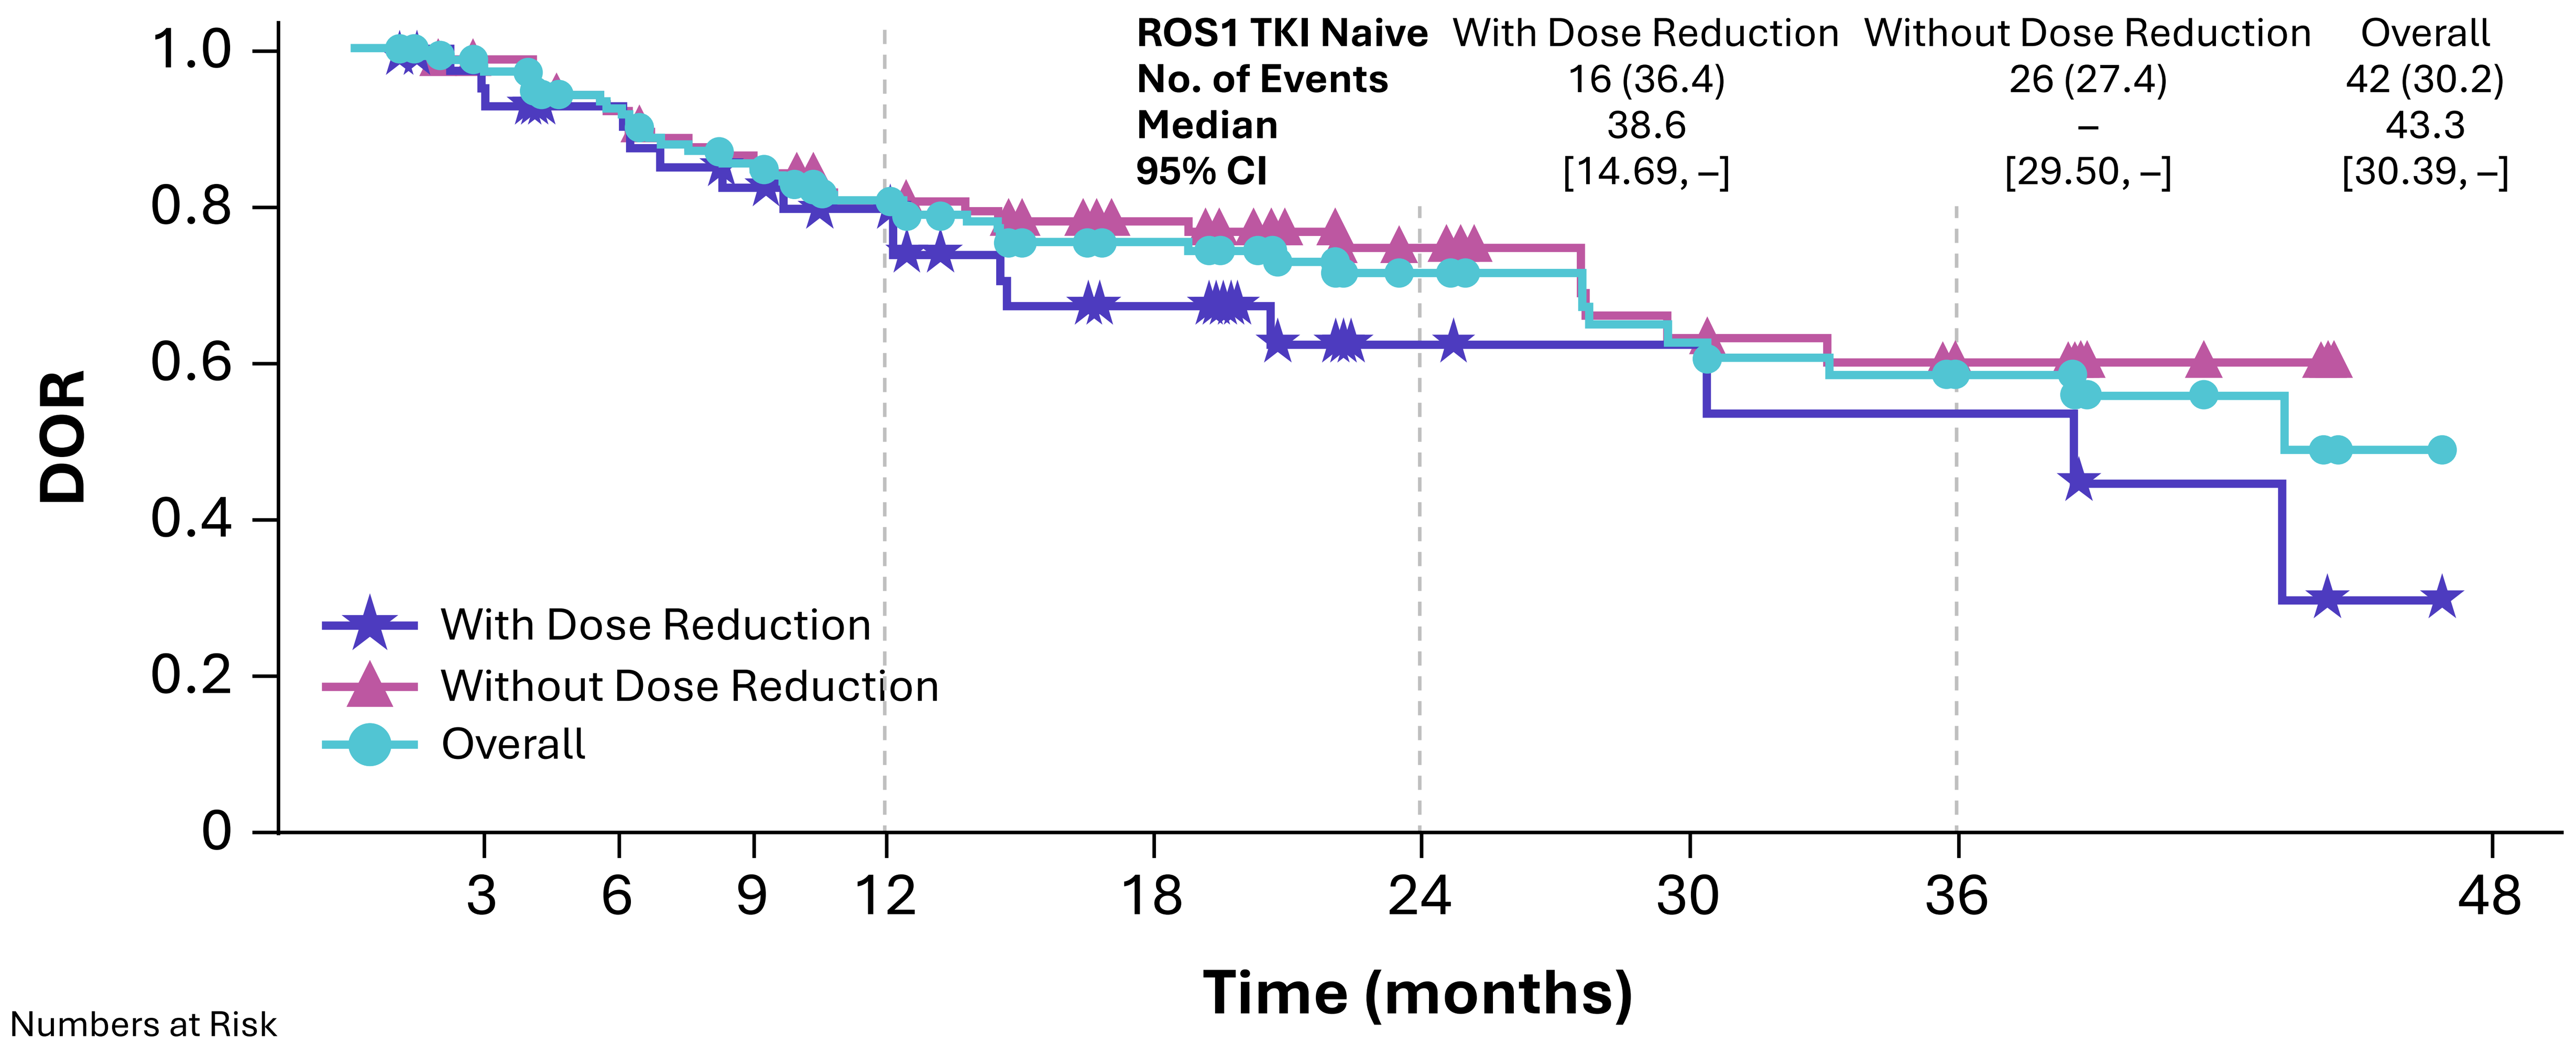
ROS1 TKI Naive | With Dose Reduction | Without Dose Reduction | Overall
No. of Events | 16 (36.4) | 26 (27.4) | 42 (30.2)
Median | 38.6 | – | 43.3
95% CI | [14.69, –] | [29.50, –] | [30.39, –]
DOR
1.0
0.8
0.6
0.4
0.2
0
3
6
9
12
18
24
30
36
48
Time (months)
With Dose Reduction
Without Dose Reduction
Overall

Numbers at Risk										
With Dose Reduction	44	39	36	31	28	19	8	7	6	0
Without Dose Reduction	95	90	83	77	68	52	29	22	18	0
Overall	139	129	119	108	96	71	37	29	24	0

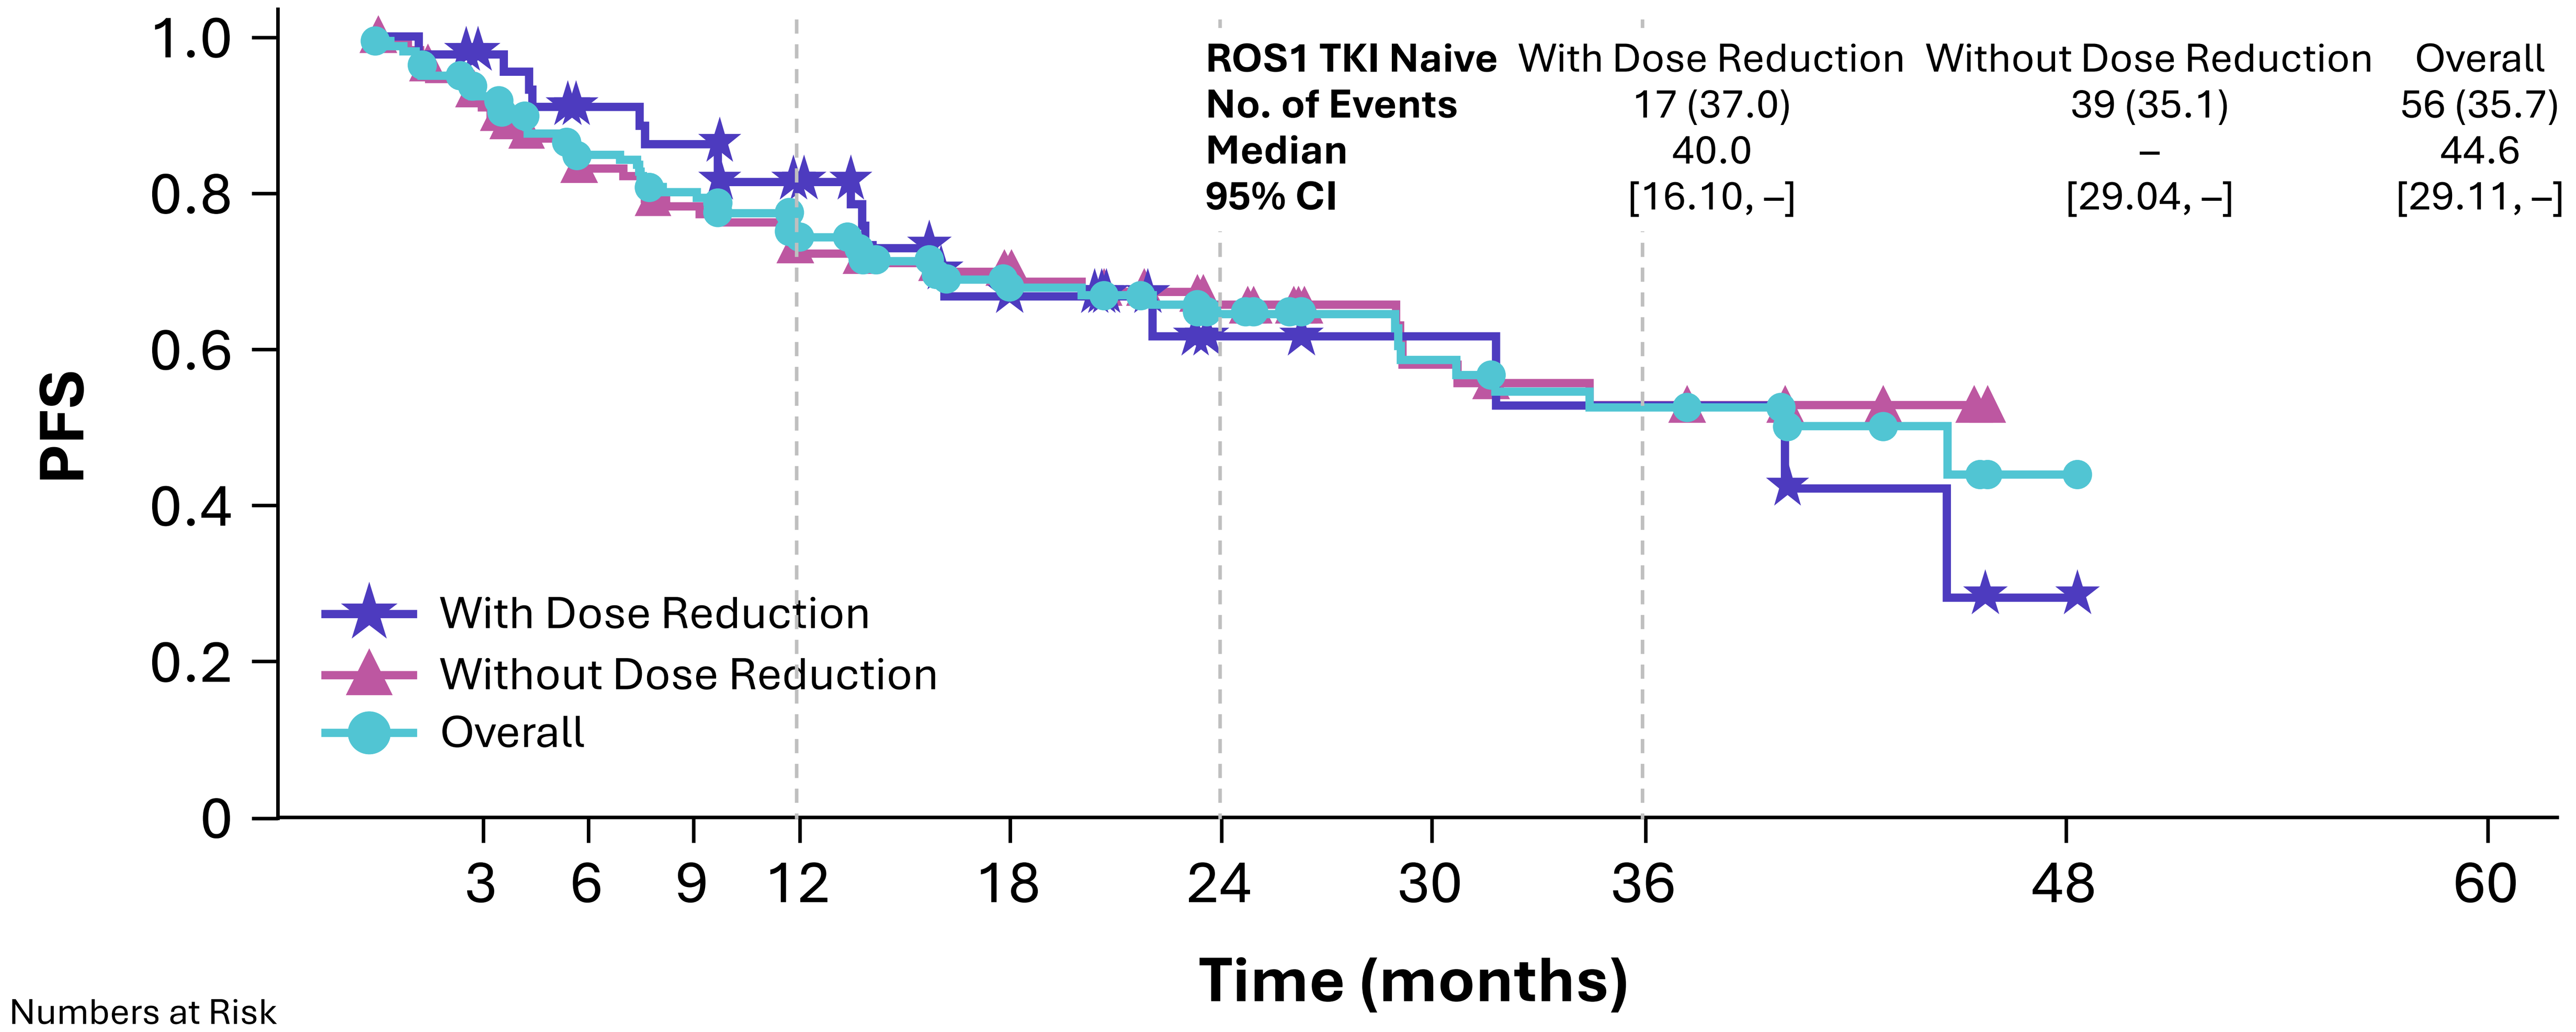
ROS1 TKI Naive | With Dose Reduction | Without Dose Reduction | Overall
No. of Events | 17 (37.0) | 39 (35.1) | 56 (35.7)
Median | 40.0 | – | 44.6
95% CI | [16.10, –] | [29.04, –] | [29.11, –]
PFS
1.0
0.8
0.6
0.4
0.2
0
3
6
9
12
18
24
30
36
48
60
Time (months)
With Dose Reduction
Without Dose Reduction
Overall

Numbers at Risk											
With Dose Reduction	46	43	38	36	31	20	8	7	6	1	0
Without Dose Reduction	111	97	85	79	70	53	31	23	20	0	0
Overall	157	140	123	115	101	73	39	30	26	1	0

TKI Pretreated

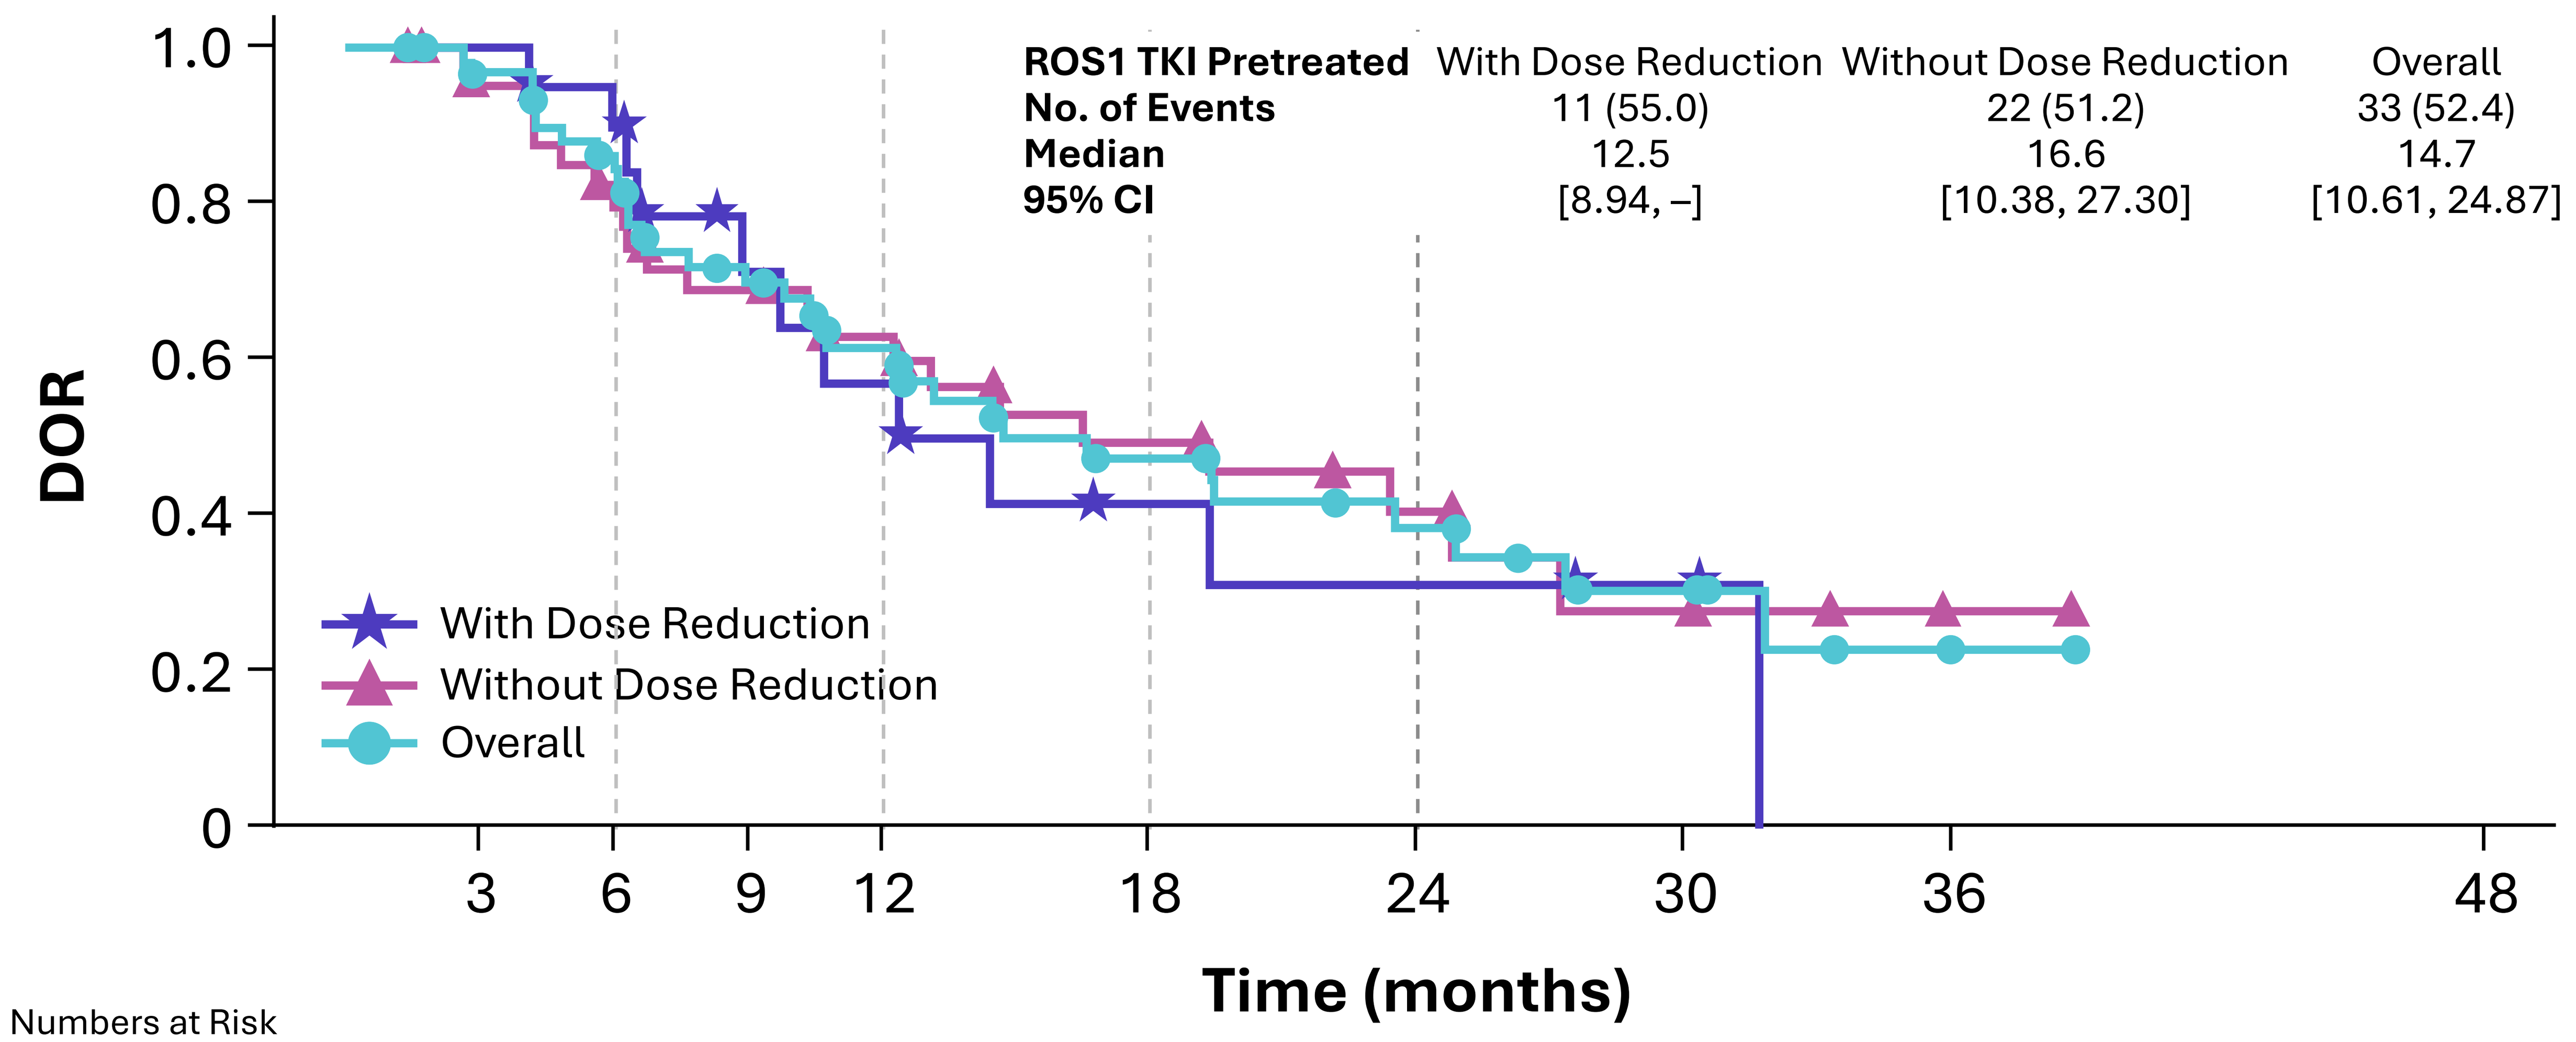
ROS1 TKI Pretreated | With Dose Reduction | Without Dose Reduction | Overall
No. of Events | 11 (55.0) | 22 (51.2) | 33 (52.4)
Median | 12.5 | 16.6 | 14.7
95% CI | [8.94, –] | [10.38, 27.30] | [10.61, 24.87]
DOR
1.0
0.8
0.6
0.4
0.2
0
3
6
9
12
18
24
30
36
48
Time (months)
With Dose Reduction
Without Dose Reduction
Overall

Numbers at Risk										
With Dose Reduction	20	20	18	10	8	4	3	2	0	0
Without Dose Reduction	43	37	31	25	20	14	8	4	1	0
Overall	63	57	49	35	28	18	11	6	1	0

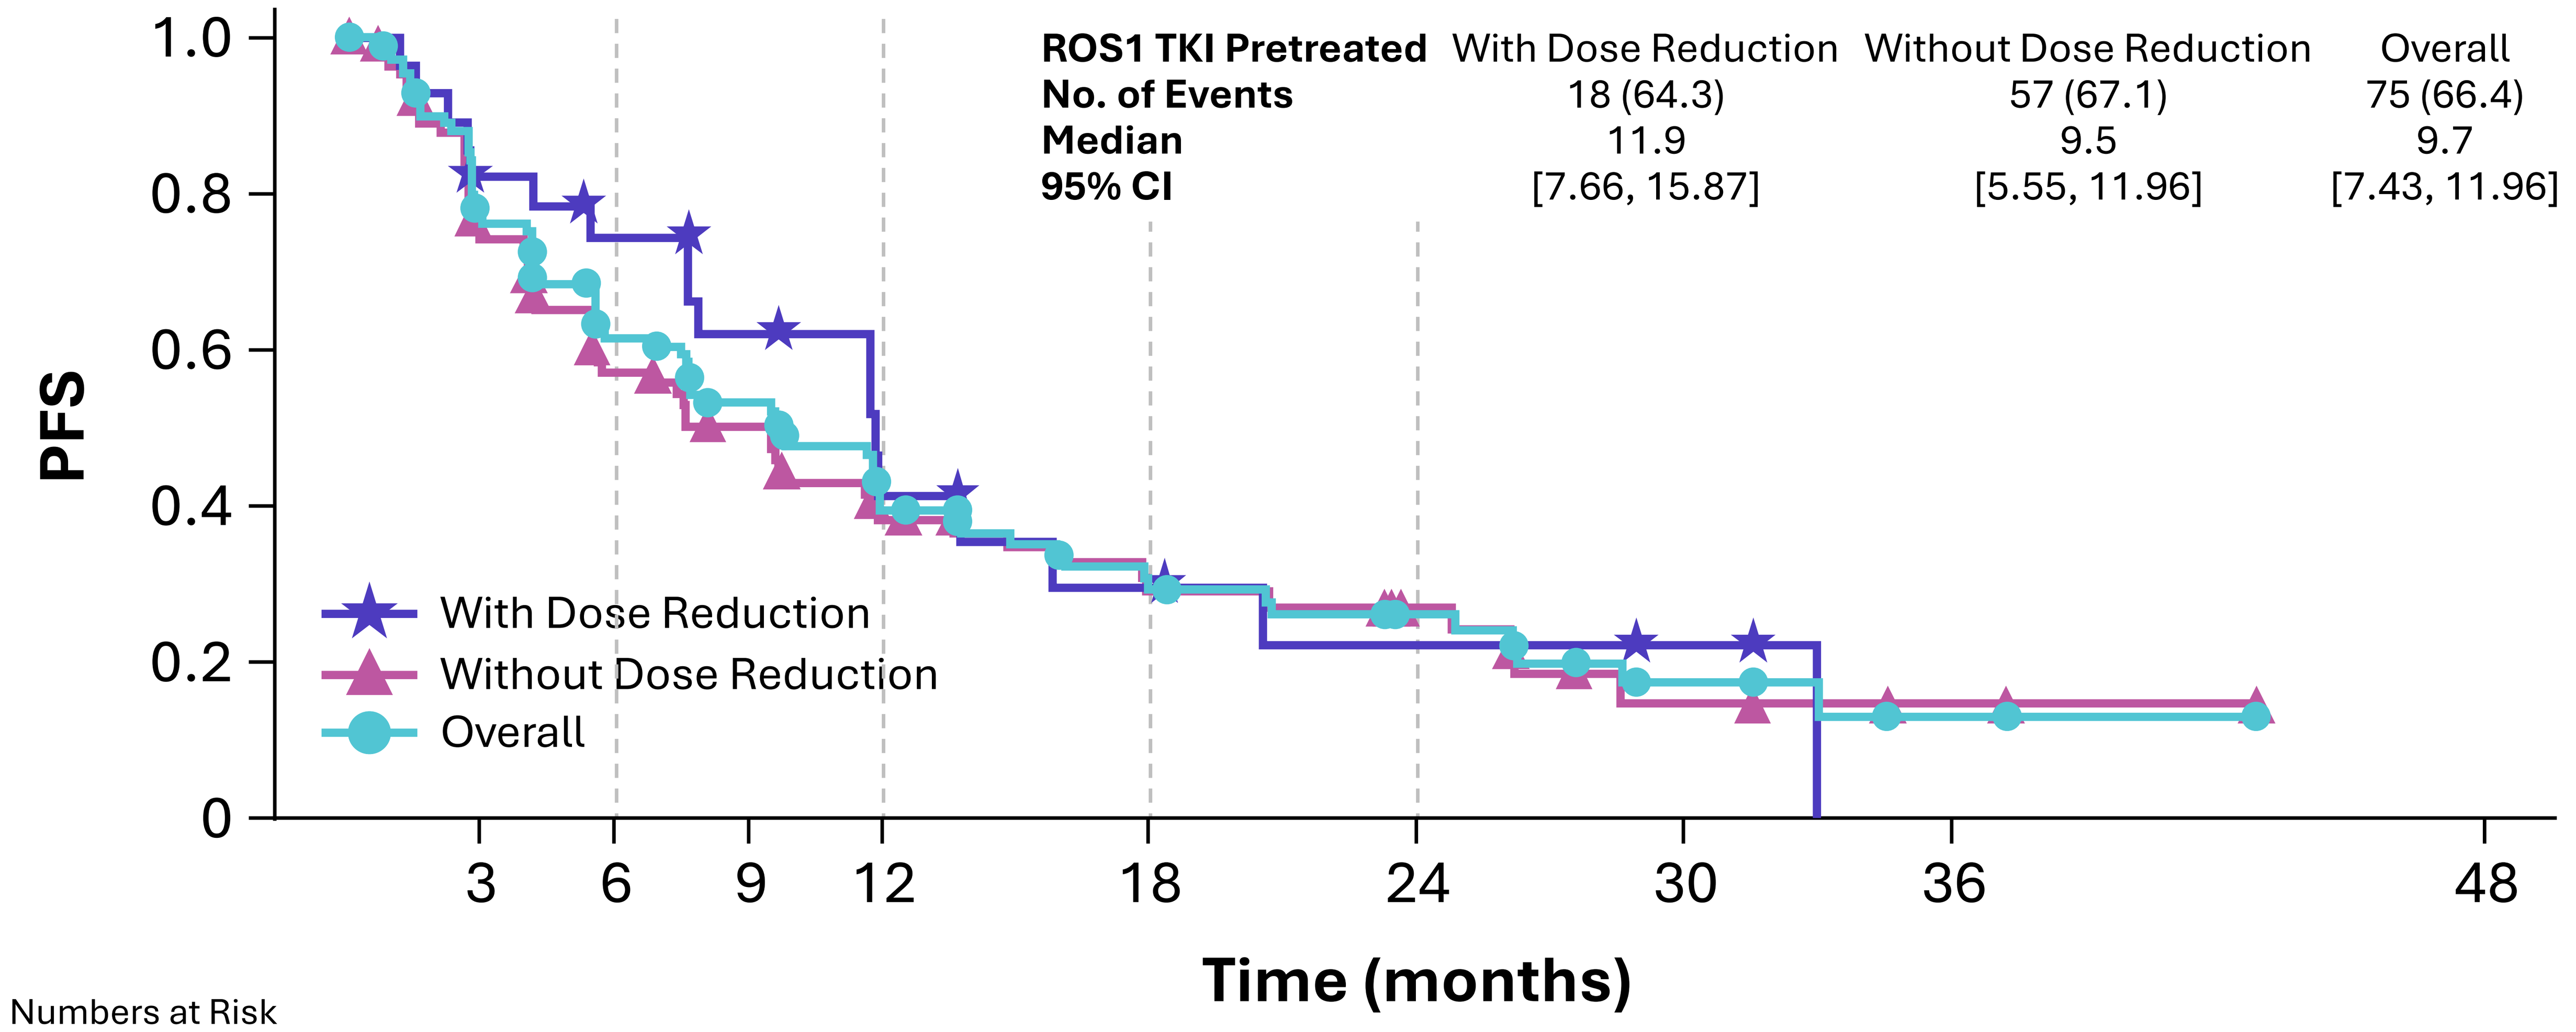
ROS1 TKI Pretreated | With Dose Reduction | Without Dose Reduction | Overall
No. of Events | 18 (64.3) | 57 (67.1) | 75 (66.4)
Median | 11.9 | 9.5 | 9.7
95% CI | [7.66, 15.87] | [5.55, 11.96] | [7.43, 11.96]
PFS
1.0
0.8
0.6
0.4
0.2
0
3
6
9
12
18
24
30
36
48
Time (months)
With Dose Reduction
Without Dose Reduction
Overall

Numbers at Risk										
With Dose Reduction	28	22	19	15	8	5	3	2	0	0
Without Dose Reduction	85	59	42	35	23	15	10	4	2	0
Overall	113	81	61	50	31	20	13	6	2	0

Data cutoff: October 28, 2024.

[a]Efficacy was assessed in the REP which included patients with advanced *ROS1*+ NSCLC with ≥1 measurable baseline lesion per RECIST v1.1 who initiated taletrectinib treatment at 600 mg QD across TKI-naive (n=157) and TKI-pretreated (n=113) cohorts. DOR, duration of response; NSCLC, non-small cell lung cancer; PFS, progression-free survival; QD, once daily; RECIST, Response Evaluation Criteria in Solid Tumors; TKI, tyrosine kinase inhibitor; REP, response evaluable population; ROS1, c-ros oncogene 1; *ROS1*+, *ROS1*-positive; TKI, tyrosine kinase inhibitor.

TRUST-II and TRUST-I: Most Grade ≥3 ALT/AST Increase Occurred Early and Were Not Cumulative[a,b]

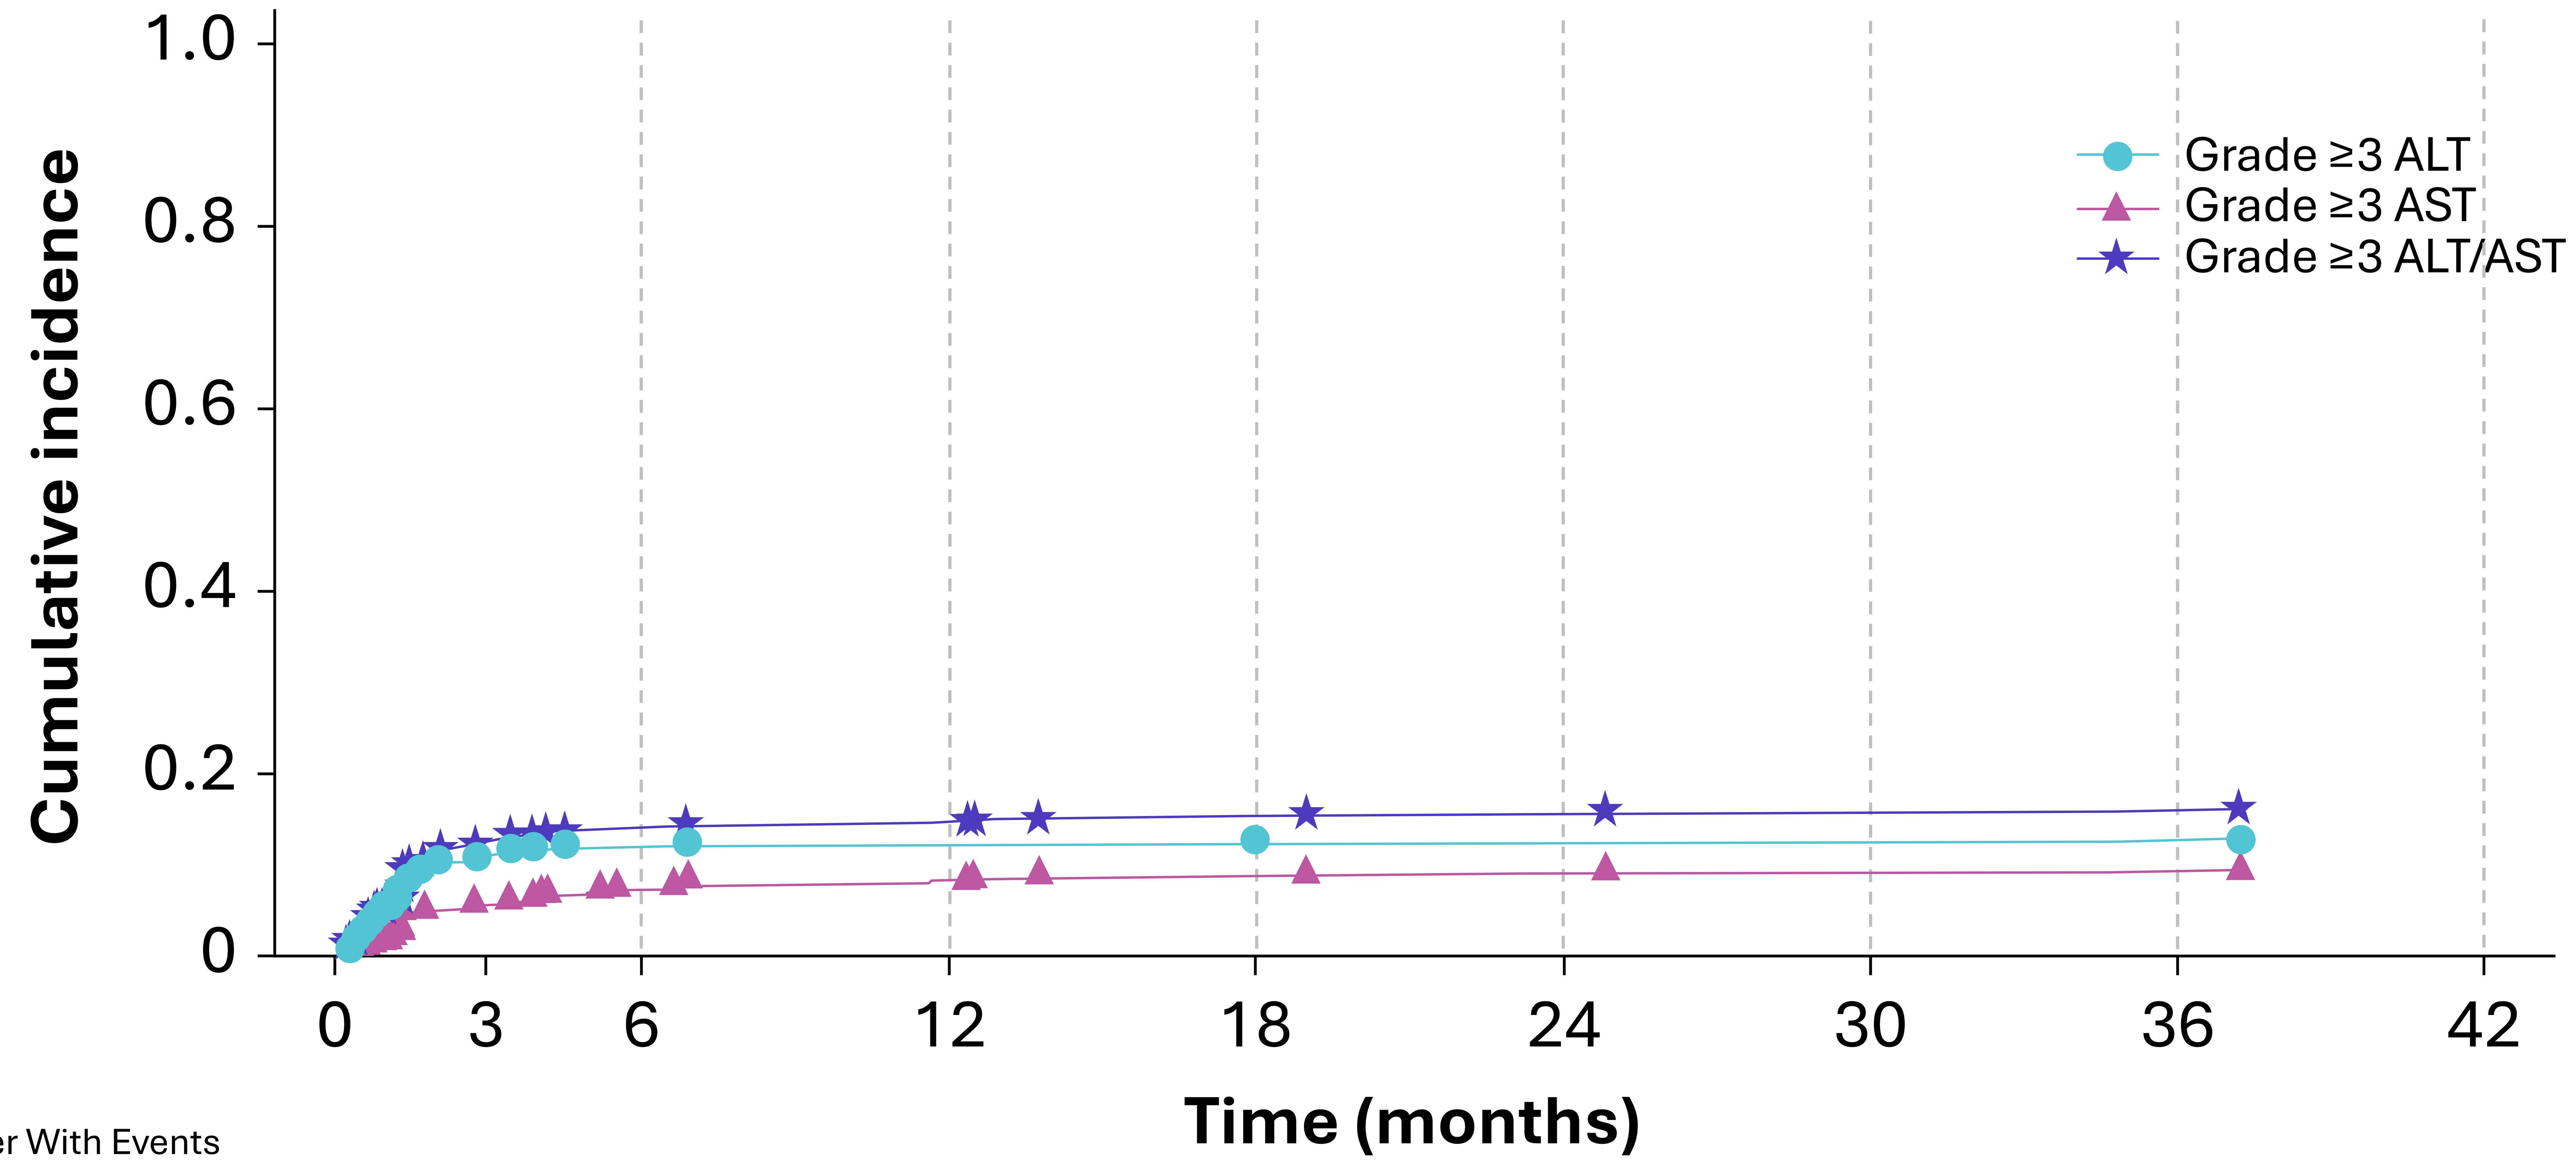
Cumulative incidence
1.0
0.8
0.6
0.4
0.2
0
0
3
6
12
18
24
30
36
42
Time (months)
Grade ≥3 ALT
Grade ≥3 AST
Grade ≥3 ALT/AST

Number With Events (Cumulative Incidents)									
Grade 3+ ALT (n=344)	0 (0.00)	38 (0.11)	42 (0.12)	43 (0.13)	43 (0.13)	44 (0.13)	44 (0.13)	44 (0.13)	45 (0.13)
Grade 3+ AST (n=344)	0 (0.00)	20 (0.06)	26 (0.08)	28 (0.08)	31 (0.09)	32 (0.09)	33 (0.10)	33 (0.10)	34 (0.10)
Grade 3+ ALT/AST (n=344)	0 (0.00)	43 (0.13)	48 (0.14)	51 (0.15)	53 (0.15)	54 (0.16)	55 (0.16)	55 (0.16)	56 (0.16)

Data cutoff: October 28, 2024.

[a]Cumulative incidence was calculated as the cumulative number of patients with first onset grade ≥3 ALT/AST evaluation events during treatment period divided by the total number of patients who had at least 1 ALT/AST laboratory assessment. [b]Safety was assessed in all patients who received ≥1 dose of taletrectinib in TRUST-II (n=171) and TRUST-I (n=173).

ALT, alanine aminotransferase; AST, aspartate aminotransferase.