#1373P

Efficacy and Safety of Taletrectinib in Patients With ROS1⁺ Non-Small Cell Lung Cancer (NSCLC): Interim Analysis of Global TRUST-II Study

Maurice Pérol[1], Nong Yang[2], Chang-Min Choi[3], Yuichiro Ohe[4], Shunichi Sugawara[5], Noriko Yanagitani[6], Geoffrey Liu[7], Filippo De Braud[8], Jorge Nieva[9], Misako Nagasaka[10], Lyudmila Bazhenova[11], Caicun Zhou[12], Enriqueta Felip[13], Xianyu Zhang[14], Shuanglian Li[14], Nathan A. Pennell[15]

[1]Léon Bérard Cancer Center, Lyon, France; [2]Hunan Cancer Hospital, Xiangya School of Medicine, Central South University, Changsha, China; [3]Asan Medical Center, University of Ulsan College of Medicine, Seoul, Republic of Korea; [4]National Cancer Center Hospital, Tokyo, Japan; [5]Sendai Kousei Hospital, Miyagi, Japan; [6]The Cancer Institute Hospital of Japanese Foundation for Cancer Research, Tokyo, Japan; [7]Princess Margaret Cancer Centre, Temerty School of Medicine, University of Toronto, Toronto, Canada; [8]University of Milan, Milan, Italy; [9]Norris Comprehensive Cancer Center, University of Southern California, Los Angeles, CA, USA; [10]University of California Irvine School of Medicine and Chao Family Comprehensive Cancer Center, Orange, CA, USA; [11]University of California San Diego Moores Cancer Center, San Diego, CA, USA; [12]Shanghai Pulmonary Hospital, Tongji University School of Medicine, Shanghai, China; [13]Vall d'Hebron University Hospital, Barcelona, Spain; [14]AnHeart Therapeutics, New York, NY, USA; [15]Cleveland Clinic Taussig Cancer Institute, Cleveland, OH, USA

Copies of this poster obtained through QR Code are for personal use only and may not be reproduced without permission from the author of this poster.

Objectives

TRUST-II is evaluating the efficacy and safety of taletrectinib in patients with advanced or metastatic ROS1⁺ NSCLC and other solid tumors

Background

- Taletrectinib is a next-generation, CNS-active, ROS1 TKI with selectivity over TRKB[1]
 - Potent against ROS1 and acquired-resistance mutations, such as G2032R[1]
 - Clinical exposures at steady state sufficient to inhibit both ROS1 and ROS1[G2032R]
- Taletrectinib previously demonstrated high overall and intracranial response rates, prolonged PFS, and activity against G2032R, and had a favorable safety profile in the regional TRUST-I study (NCT04395677)[2]
 - Based on these findings, taletrectinib was granted a breakthrough therapy designation by the FDA for treatment of adults with advanced or metastatic ROS1⁺ NSCLC who are ROS1 TKI treatment-naive or previously treated with crizotinib
- Here, we present the interim results from the global pivotal phase 2 trial, TRUST-II (NCT04919811), evaluating taletrectinib in patients with advanced ROS1⁺ NSCLC from North America, Europe, and Asia[1]

Abbreviations

AE, adverse event; AST, aspartate aminotransferase; ALT, alanine aminotransferase; C, crizotinib CI, confidence interval; CNS, central nervous system; CPK; creatinine phosphokinase; CT, chemotherapy; cORR, confirmed objective response rate; cCR, confirmed complete response; cPR, confirmed partial response; DCR, disease control rate; DoR, duration of response; E, entrectinib ECG; electrocardiogram; ECOG PS, Eastern Cooperative Oncology Group performance status; IC, intracranial; IQR, interquartile range; IRC, independent review committee; mDoR, median duration of response; mPFS, median progression-free survival; NR, not reached; NSCLC, non small cell lung cancer; OS, overall survival; PD, progressive disease; PFS, progression-free survival; QD, once daily; RECIST, Response Evaluation Criteria in Solid Tumors; SD, stable disease; TEAE, treatment-emergent adverse event; TKI, tyrosine kinase inhibitor; TRKB, tropomyosin receptor kinase B; TTR, time to response.

References

1. Nagasaka M, et al. *Future Oncol.* 2023;19(2):123-135.
2. Li W, et al. *J Thorac Oncol.* 2023;S47.

Acknowledgments

On behalf of all the authors, we would like to thank the patients, study investigators, and site personnel for their participation in this study

This study was sponsored by AnHeart Therapeutics, Inc. Medical writing and editorial assistance was provided by Peloton Advantage, LLC, an OPEN Health company, and funded by AnHeart Therapeutics, Inc.

Methods

- TRUST-II (NCT04919811), a global phase 2, multicenter, open-label, single-arm study in patients with ROS1⁺ tumors, has 4 cohorts

TRUST II (NCT04919811) Study Design

Key Eligibility Criteria (N≈154)

Inclusion Criteria:

- Locally advanced or metastatic NSCLC or other solid tumors
- Age ≥18 years
- ECOG PS 0–1
- Evidence of ROS1 fusion
- Stable CNS involvement allowed (including leptomeningeal carcinomas)
- At least 1 measurable lesion per RECIST v1.1

Cohort 1: NSCLC: ROS1 TKI-naive, ≤1 CT

Cohort 2[a]: NSCLC: 1 prior ROS1 TKI, ≤1 line of CT

Cohort 3[b]: NSCLC: ≥2 prior ROS1 TKIs, ≤1 line of CT

Cohort 4[b]: Solid tumors including NSCLC if ineligible for cohorts 1–3; ≤3 ROS1 TKIs, ≤2 lines of CT

Taletrectinib[c] 600 mg QD, oral

Study Endpoints

Primary:
- IRC-assessed cORR per RECIST v1.1

Key Secondary:
- Intracranial cORR (RECIST v1.1)
- DoR
- DCR
- TTR (RECIST v1.1)
- PFS
- OS
- Safety

Study Populations:
- Safety – All patients who received ≥1 dose of taletrectinib
- Efficacy[d] – Patients with documented ROS1 fusion, ≥1 measurable lesion at baseline per IRC, and approximately 6 months of follow-up

[a]Prior TKI use was limited to approved ROS1 TKIs (crizotinib or entrectinib). [b]Prior TKI use could include any ROS1 TKI regardless of approval status, per most recent protocol amendment 3.0. [c]Taletrectinib was administered in 21-day cycles. [d]For interim analysis only.

Results

- Data cutoff date: July 12, 2023
- The efficacy population presented here includes patients from cohorts 1 and 2

Patient Demographics and Baseline Characteristics (Efficacy Population)

46 patients enrolled in cohorts 1 and 2 with ~6 months of follow-up prior to July 12, 2023[a]

Median age: **58 y**
IQR: 52.8-67.0

21 (46%) of patients were female

Smoking status

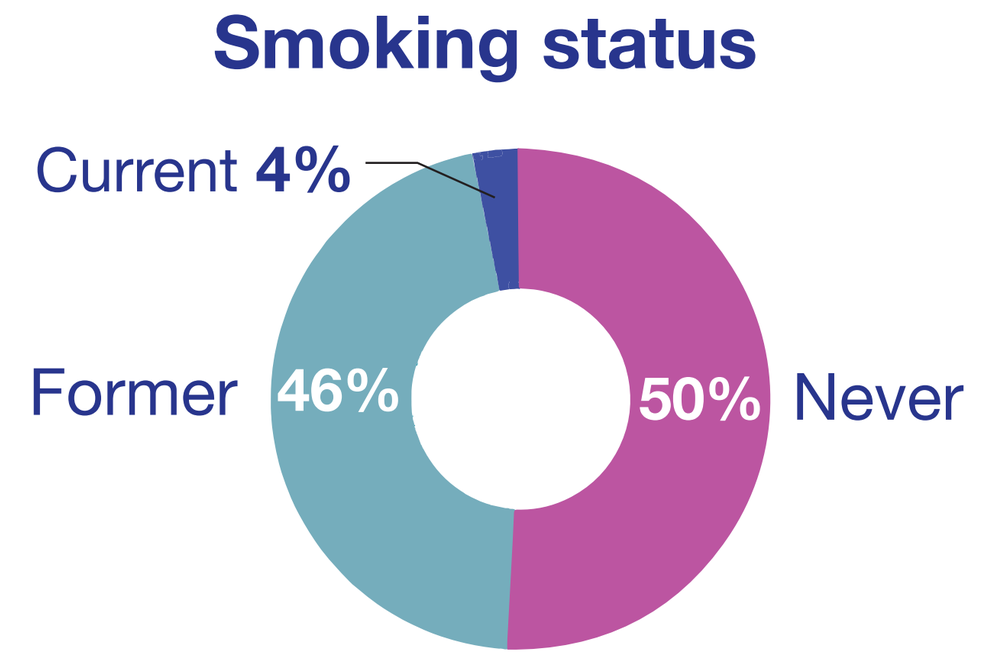
Current 4%
Former 46%
50% Never

ECOG PS

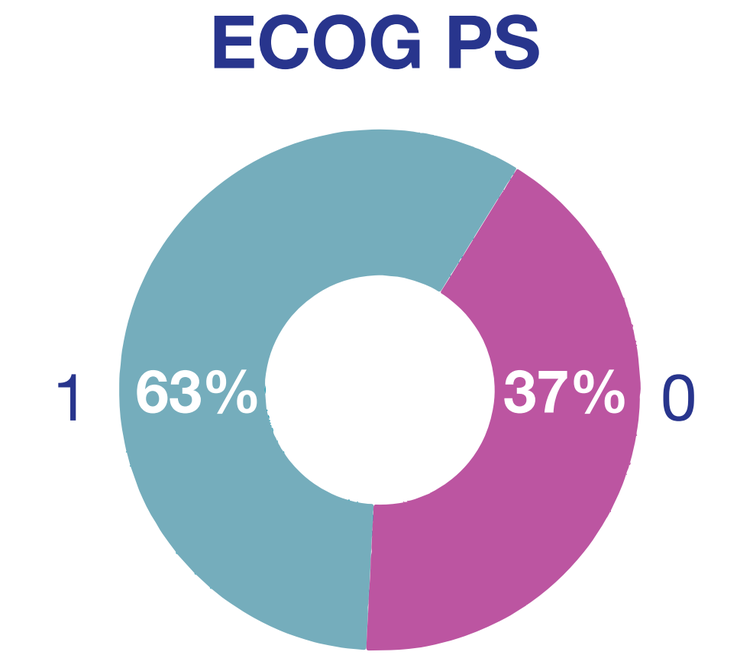
1 63%
37% 0

Prior chemotherapy

TKI-Naive 16%

TKI-pretreated 52%

Brain metastasis[b,c]

TKI-Naive 32%

TKI-pretreated 52%

[a]The efficacy population consisted of 25 TKI-Naive patients in cohort 1 and 21 TKI-pretreated patients in cohort 2.
[b]Brain metastasis status by IRC (RANO-BM). [c]25% (cohort 1) and 73% (cohort 2) of patients with baseline brain metastases had prior radiotherapy to the brain.

Responses by Cohort[a,b]

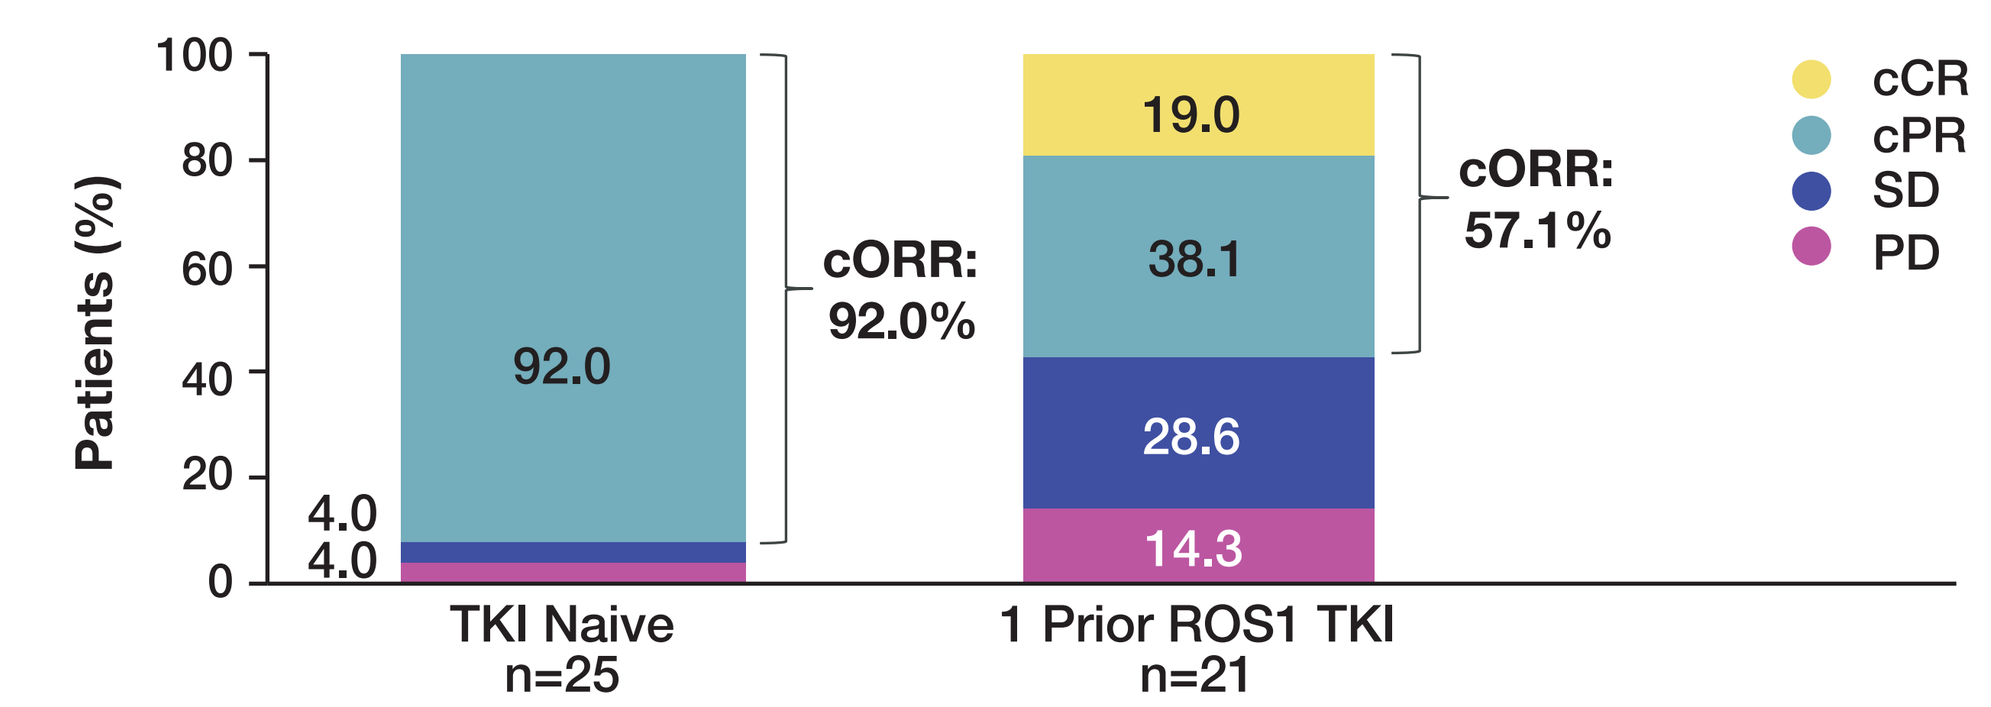
Patients (%)
100
80
60
40
20
0
92.0
4.0
4.0
cORR: 92.0%
19.0
38.1
28.6
14.3
cORR: 57.1%
TKI Naive n=25
1 Prior ROS1 TKI n=21
cCR
cPR
SD
PD

	TKI-Naive (n=25)	TKI-pretreated (n=21)
Median follow-up, months (95% CI)	8.1 (5.6, 11.7)	11.7 (5.5, 15.8)
Median PFS, months (95% CI)	NR (12.0, NR)	11.7 (4.0, NR)
12-month PFS rate, % (95% CI)	82.3 (59.2, 93.0)	42.6 (18.0, 65.5)

[a]Efficacy population. [b]Assessed by an independent review committee per RECIST v1.1.

- Duration of response was not mature in either cohort

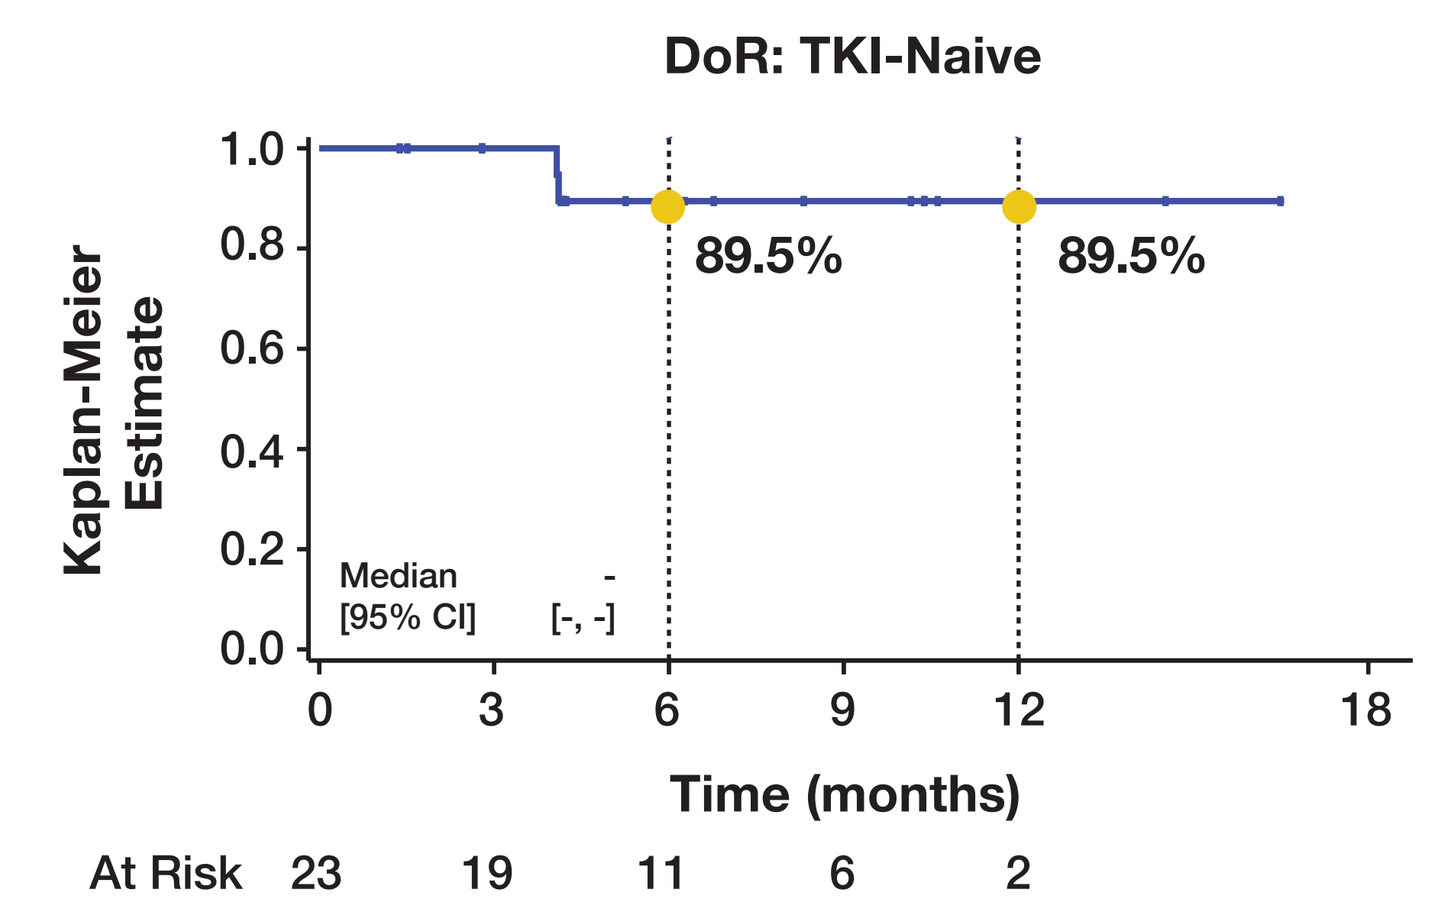
DoR: TKI-Naive
Kaplan-Meier Estimate
89.5%
89.5%
Median [95% CI] - [-, -]
Time (months)
At Risk 23 19 11 6 2

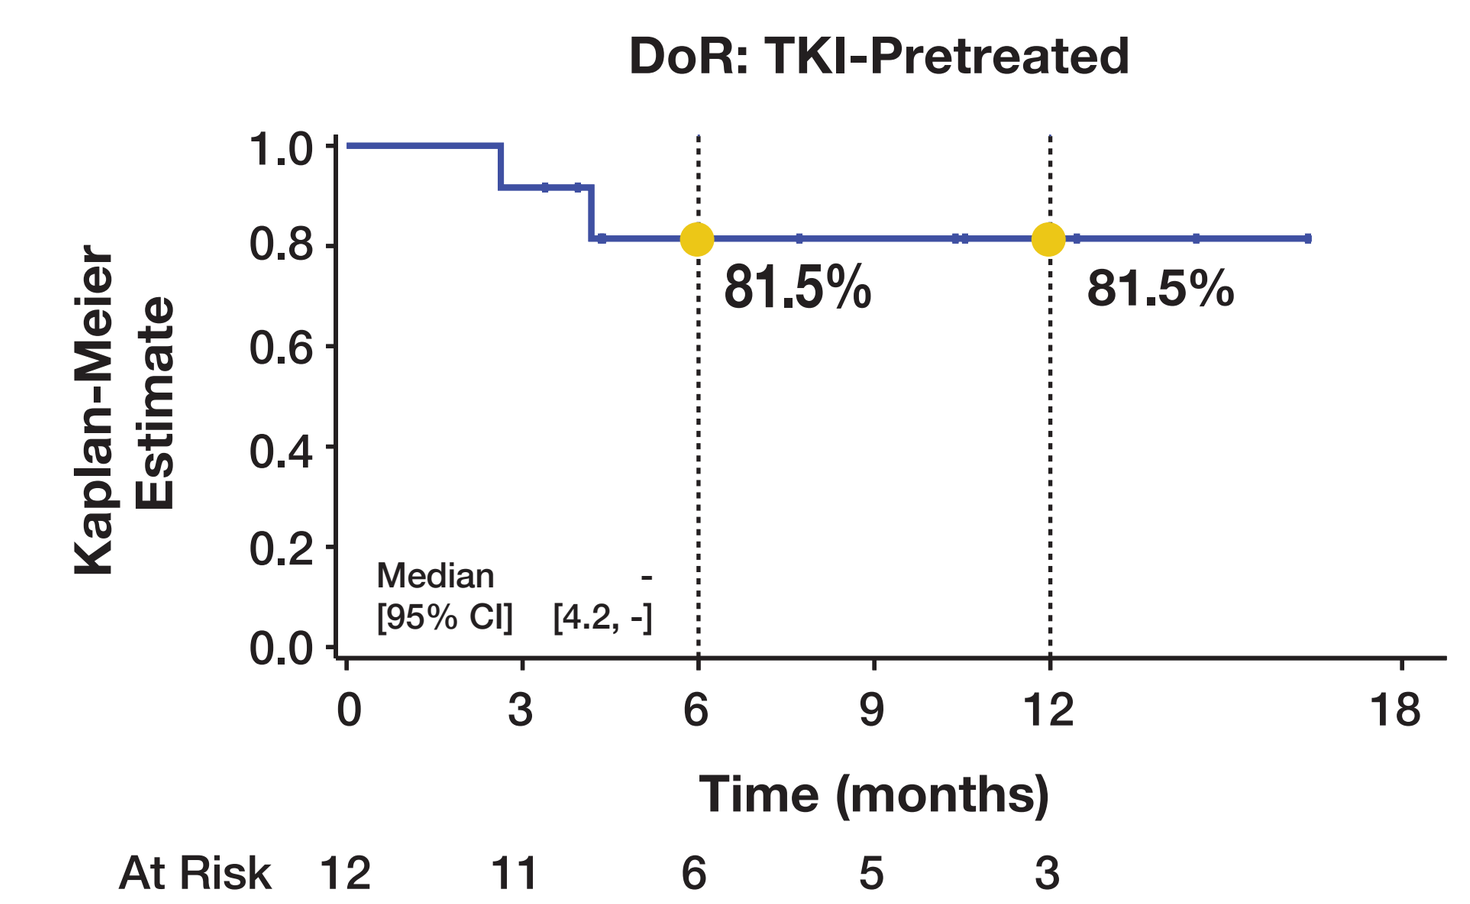
DoR: TKI-Pretreated
Kaplan-Meier Estimate
81.5%
81.5%
Median [95% CI] - [4.2, -]
Time (months)
At Risk 12 11 6 5 3

Efficacy in Patients With Brain Metastases[a]

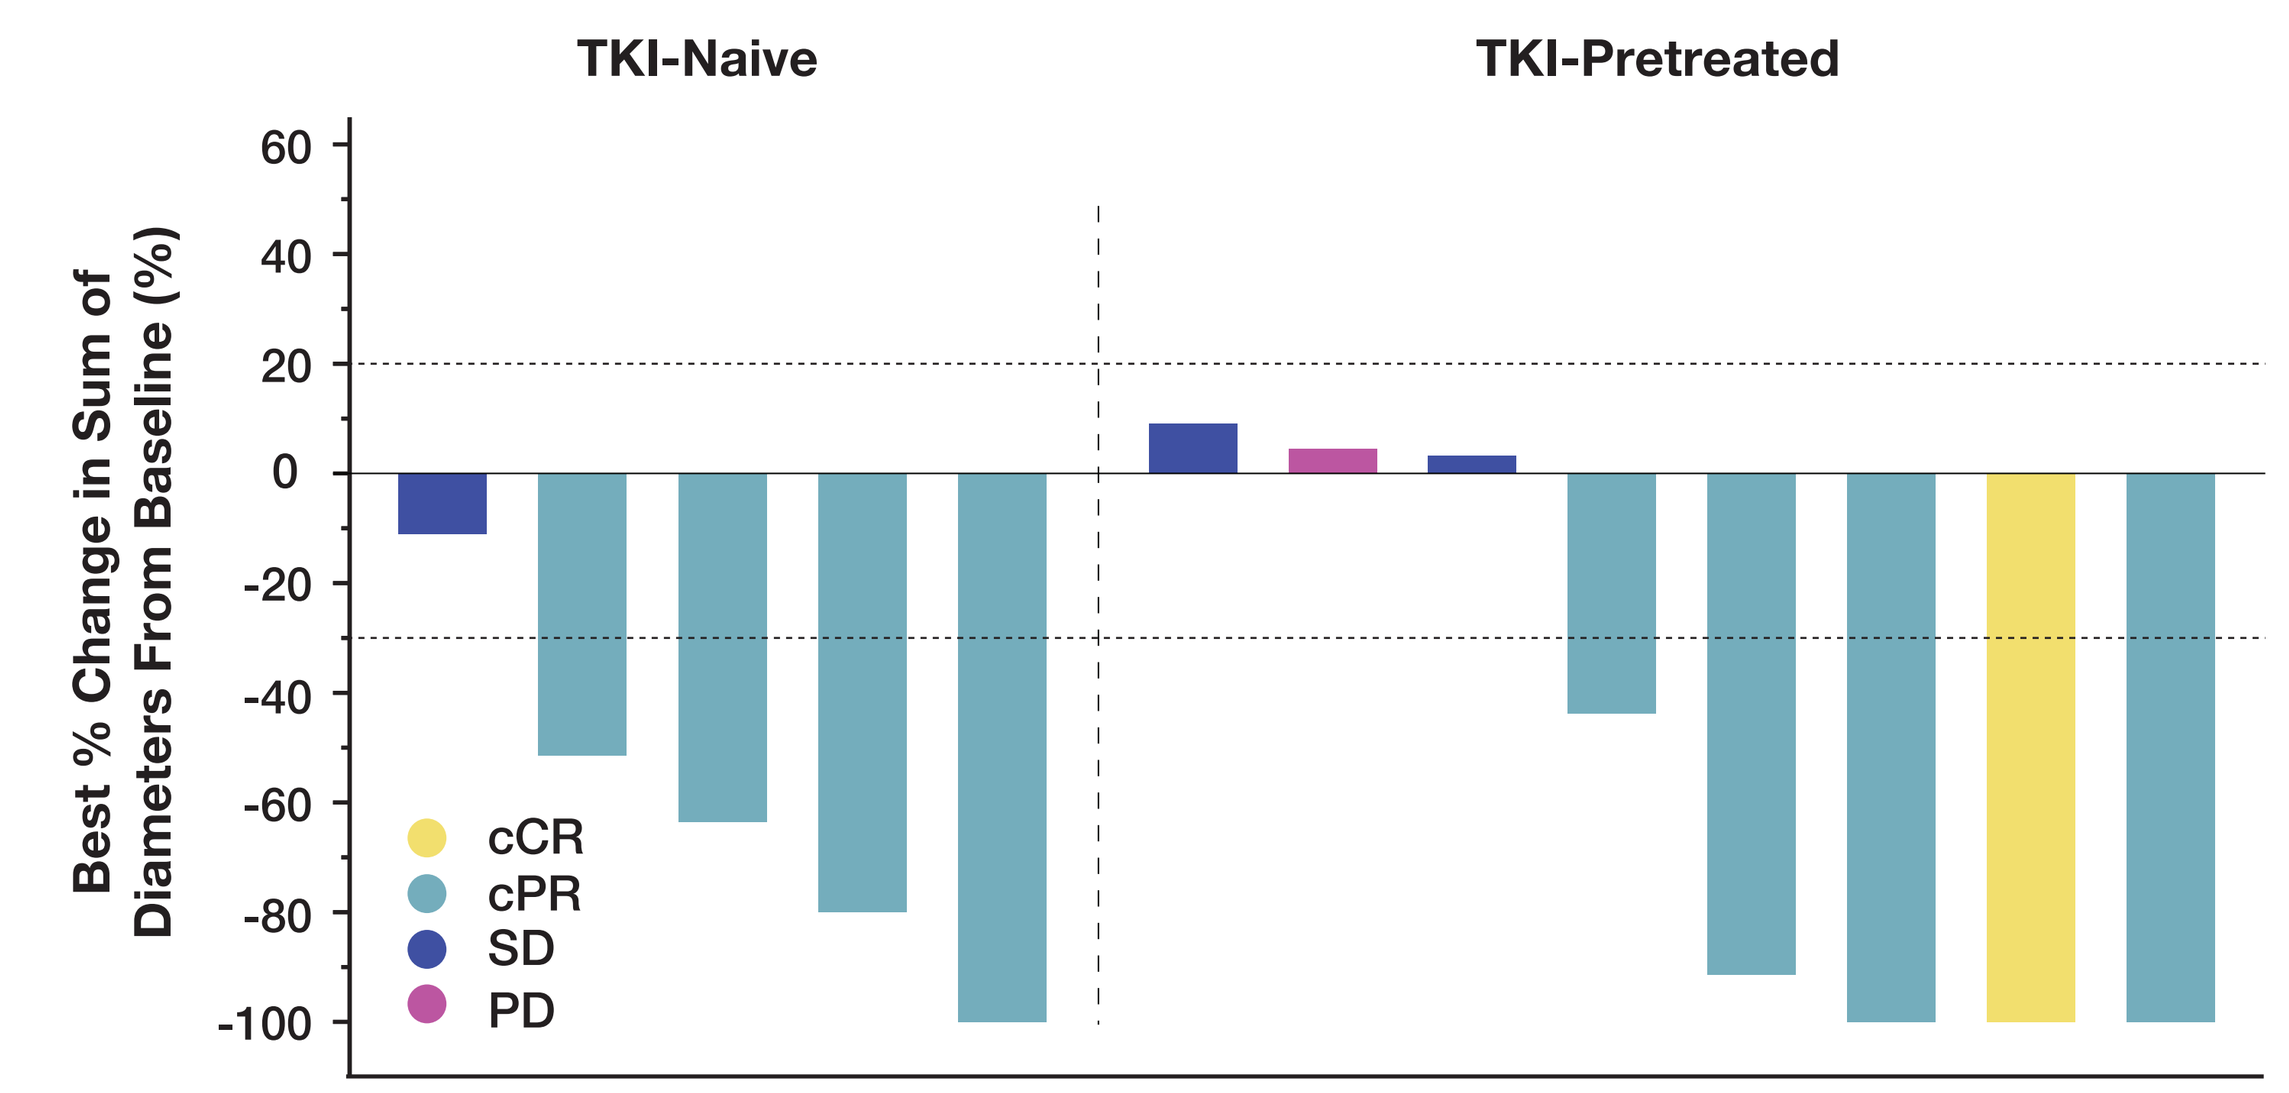
TKI-Naive
TKI-Pretreated
Best % Change in Sum of Diameters From Baseline (%)
cCR
cPR
SD
PD

	TKI-Naive (n=5)	TKI-pretreated (n=8)
IC-cORR, % (n/N) [95% CI]	80.0 (4/5) [28.4, 99.5]	62.5 (5/8) [24.5, 91.5]

[a]Measurable lesions per RECIST v1.1.

Summary

- Interim analysis from the global pivotal TRUST-II study further establishes taletrectinib as a promising new option for patients with ROS1⁺ NSCLC
 - Tumor response was robust, durable, and consistent with TRUST-I
 - High overall response rates were observed in both TKI-naive (92%) and TKI-pretreated patients (57%)
 - Intracranial responses were robust in both TKI-naive (80%) and TKI-pretreated patients (63%)
- Taletrectinib demonstrated a favorable safety profile in ROS1 TKI-naive and TKI-pretreated NSCLC patients
 - TEAEs were mostly grade 1–2; incidence of dizziness was low (13%)
 - Low incidence of treatment discontinuations due to TEAEs
 - No treatment-related deaths
- Enrollment in this global phase 2 trial (NCT04919811) of taletrectinib is currently ongoing

Efficacy in ROS1 TKI-Naive Patients (TKI-Naive; n=25)

- cORR: 92.0% (95% CI: 74.0, 99.0)

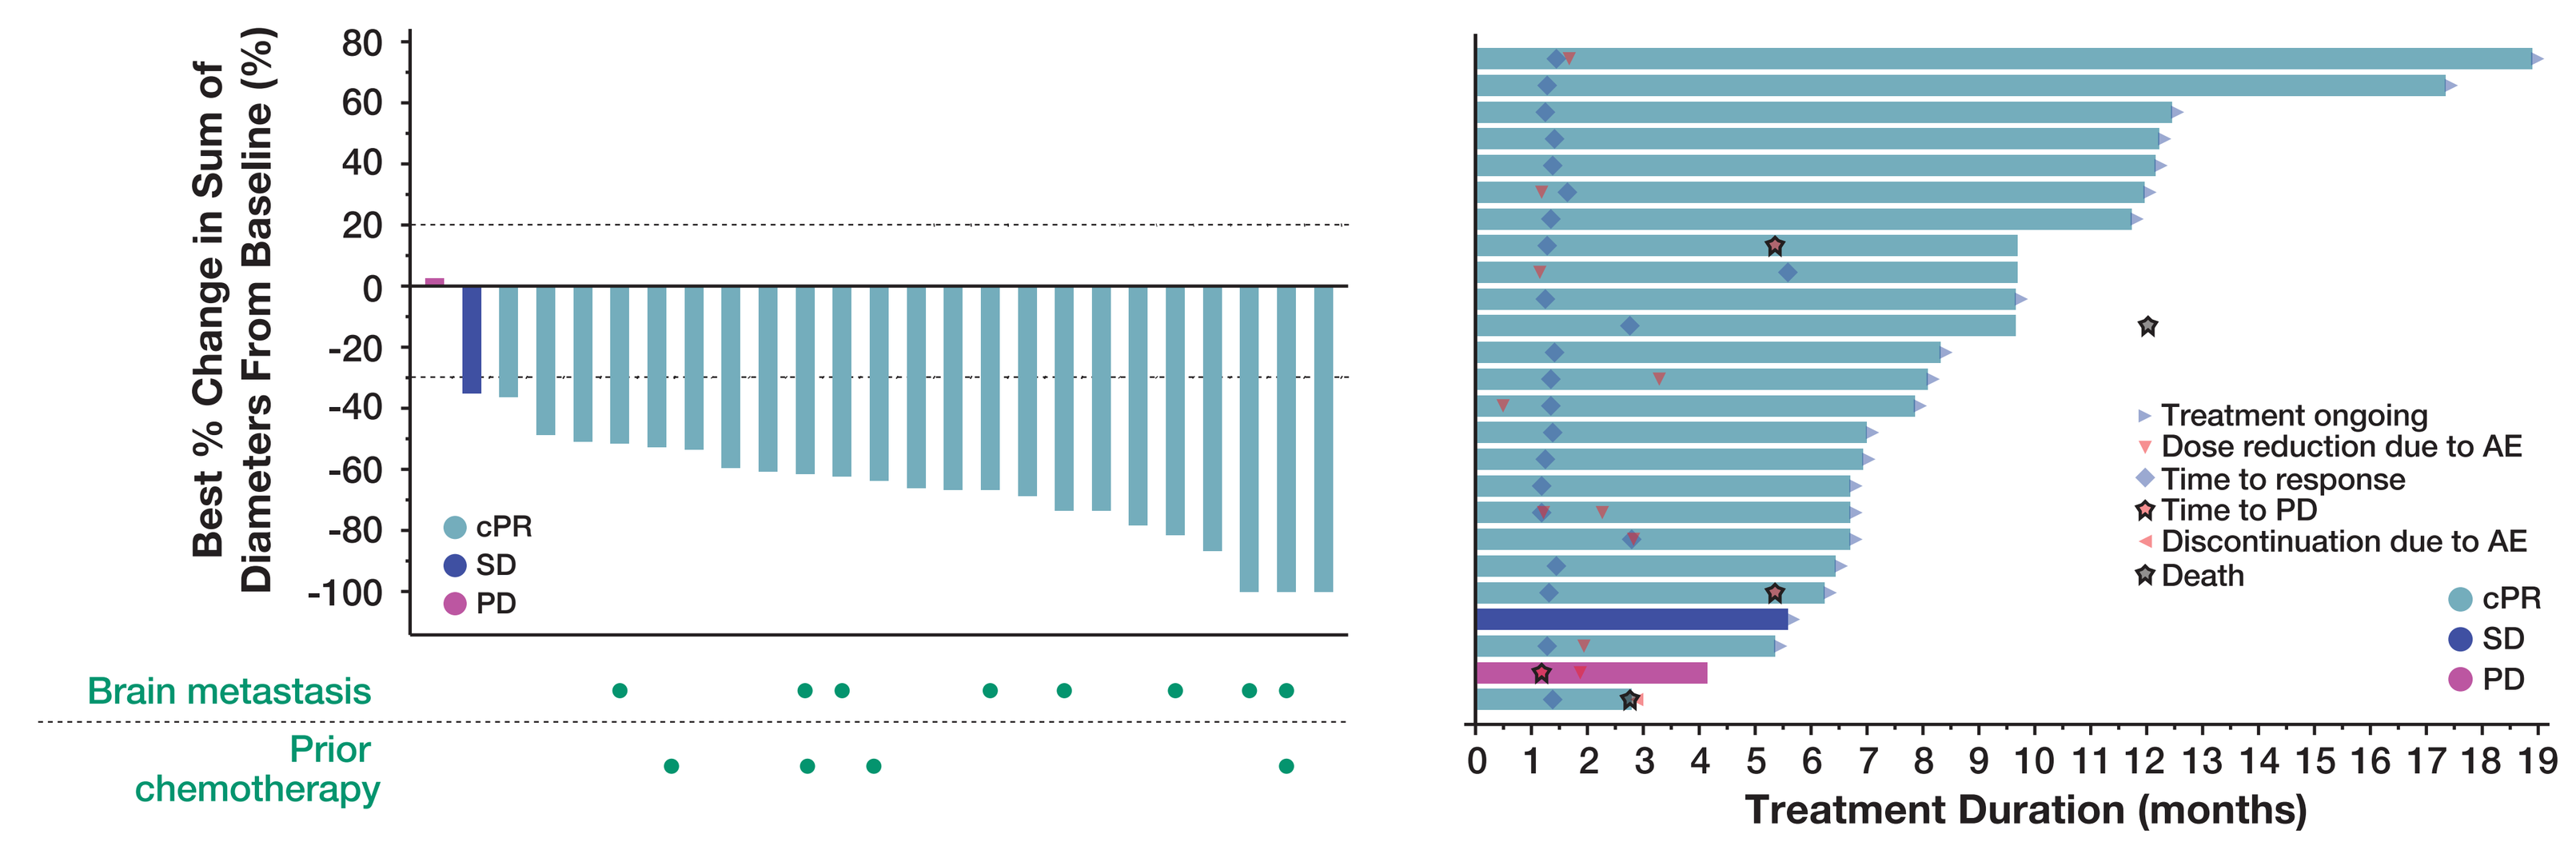
Best % Change in Sum of Diameters From Baseline (%)
cPR
SD
PD
Brain metastasis
Prior chemotherapy
Treatment ongoing
Dose reduction due to AE
Time to response
Time to PD
Discontinuation due to AE
Death
Treatment Duration (months)

Efficacy in ROS1 TKI-Pretreated Patients (TKI-pretreated; n=21)

- cORR: 57.1% (95% CI: 34.0, 78.2)
- 17 patients (81%) in Cohort 2 received crizotinib (C); 4 (19%) received entrectinib (E)

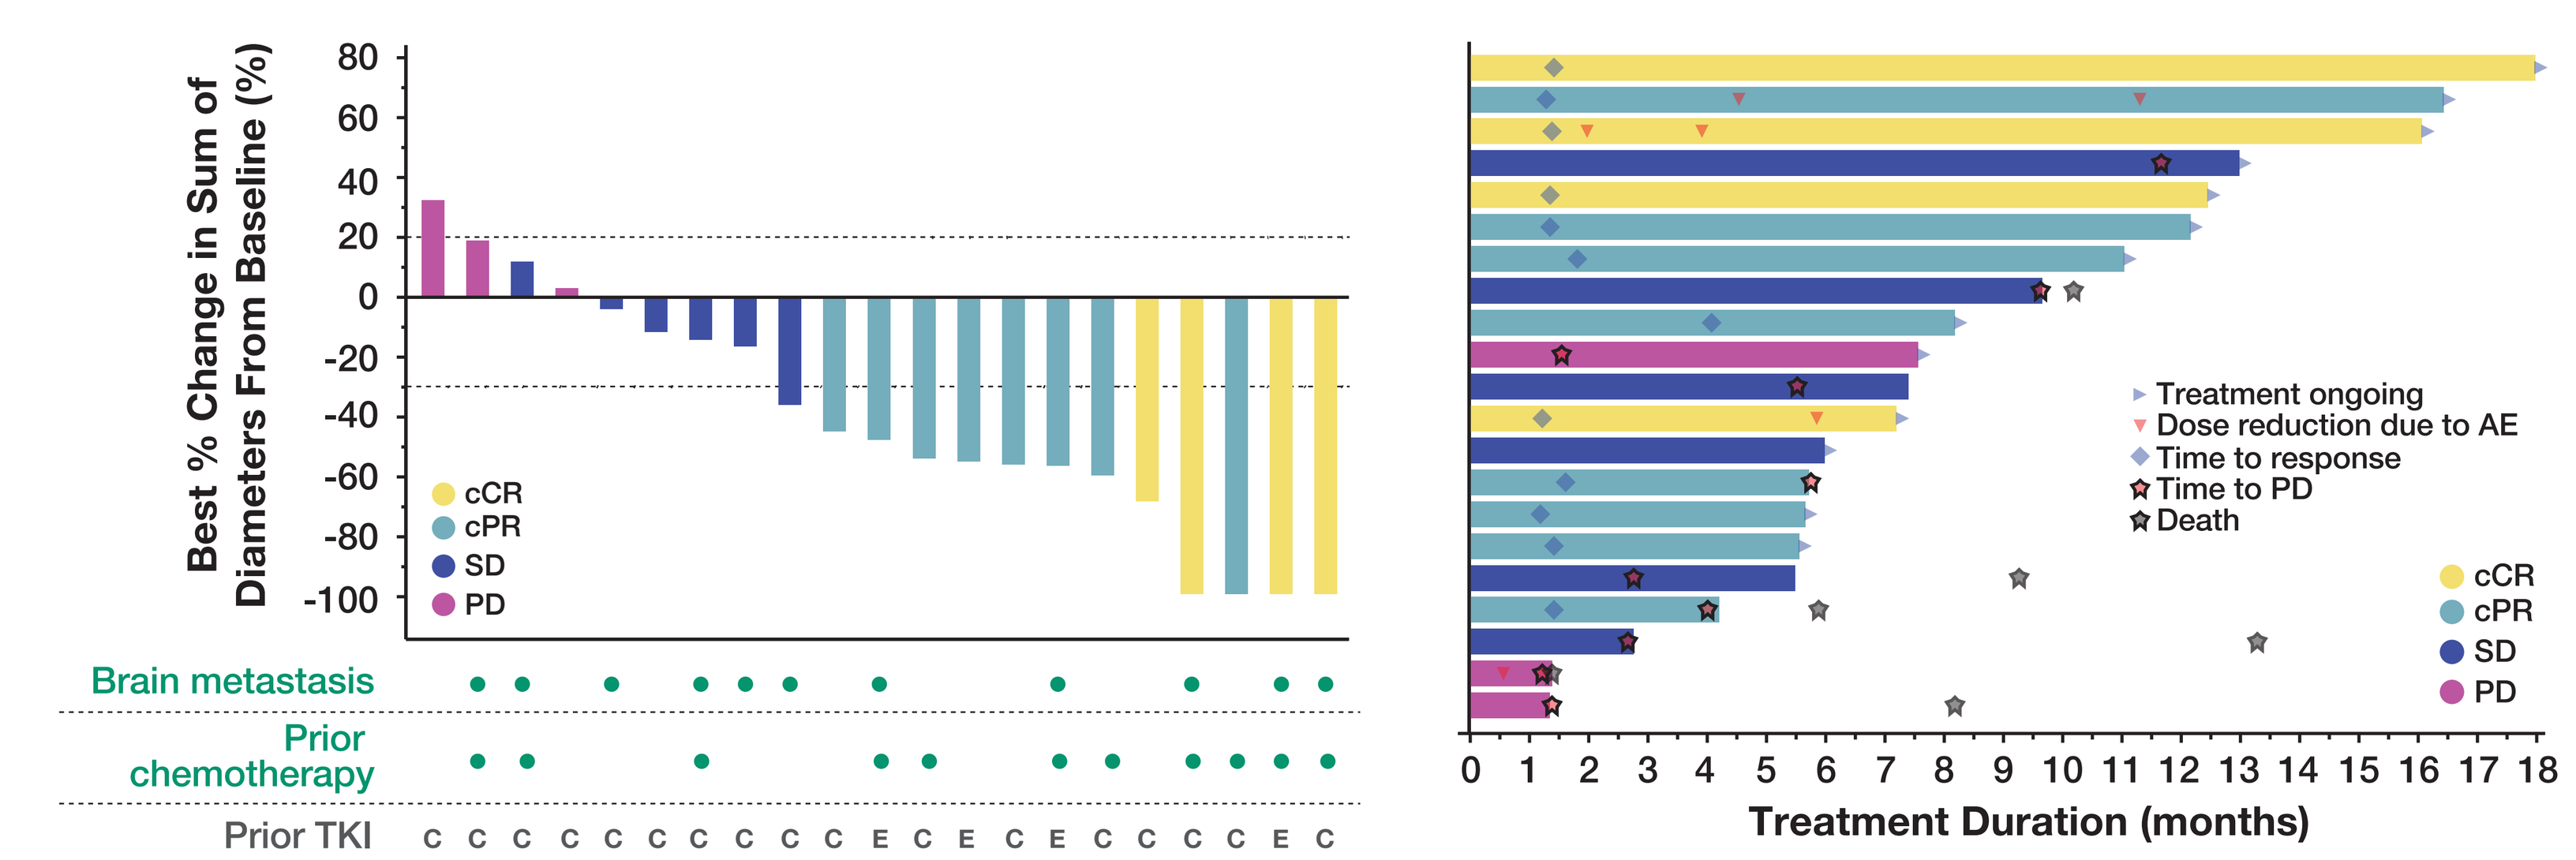
Best % Change in Sum of Diameters From Baseline (%)
cCR
cPR
SD
PD
Brain metastasis
Prior chemotherapy
Prior TKI
C C C C C C C C C E C E C E C C C C E C
Treatment ongoing
Dose reduction due to AE
Time to response
Time to PD
Death
Treatment Duration (months)

Safety (Safety Population, n=107)[a]

- Median duration of exposure was 4.9 months (range: 0.09–18.9)
- Most common TEAEs: increased ALT (64%) and AST (63%), diarrhea (43%), and nausea (43%)
- Most TEAEs were grade 1–2
- 12% of patients had serious AEs
- Neurological TEAEs were low, majority were grade 1–2
 - 13% of patients experienced dizziness; most were grade 1
- TEAEs led to dose reduction in 34% of patients, the most common was increased liver enzymes (23%)
- 2% of patients had a TEAE leading to treatment discontinuation
- No taletrectinib-related deaths were reported

Patients With TEAEs (≥15%): Taletrectinib (Safety Population; n=107)[a]

TEAE, n (%)	Grade 1	Grade 2	Grade 3	Grade 4	Grade 5	All Grade
ALT increased	37 (34.6)	14 (13.1)	17 (15.9)	0	0	68 (63.6)
AST increased	40 (37.4)	23 (21.5)	4 (3.7)	0	0	67 (62.6)
Diarrhea	37 (34.6)	8 (7.5)	1 (0.9)	0	0	46 (43.0)
Nausea	32 (29.9)	11 (10.3)	3 (2.8)	0	0	46 (43.0)
Vomiting	23 (21.5)	7 (6.5)	1 (0.9)	0	0	31 (29.0)
Dysgeusia	13 (12.1)	7 (6.5)	0	0	0	20 (18.7)
Anemia	11 (10.3)	4 (3.7)	3 (2.8)	0	0	18 (16.8)
Constipation	15 (14.0)	3 (2.8)	0	0	0	18 (16.8)
Blood CPK increased	10 (9.3)	2 (1.9)	4 (3.7)	0	0	16 (15.0)

[a]Worst grade per patient is reported.

Presented at the European Society for Medical Oncology (ESMO) Annual Meeting I October 20–24, 2023 I Madrid, Spain

Supplemental materials

Table S1. Patient Demographics and Baseline Characteristics (Efficacy Population)

Key Demographics	TKI-Naive (n=25) (%)	TKI-Pretreated (n=21) (%)
Age, median (range), y	60.0 (39–77)	55.0 (38–77)
Female, n (%)	11 (44.0)	10 (47.6)
Western/Asian region, n (%)	6 (24.0)/19 (76.0)	6 (28.6)/15 (71.4)
ECOG PS 0/1, n (%)	11 (44.0)/14 (56.0)	6 (28.6)/15 (71.4)
Prior chemotherapy, n (%)	4 (16.0)	11 (52.4)
Prior crizotinib/entrectinib, n (%)	0	7 (80.9)/4 (19.0)
Never/former/current smoker, n (%)	11 (44.0)/13 (52.0)/1 (4.0)	12 (57.1)/8 (38.1)/1 (4.8)
Brain metastasis, n (%)	8 (32.0)	11 (52.4)
Previously treated with radiotherapy, n (%)	2 (25.0)	8 (72.3)
Previously treated with stereotactic radiosurgery, n (%)	2 (25.0)	7 (63.6)
Previously treated with whole brain radiotherapy, n (%)	0	1 (9.1)

AnHeart Therapeutics

Table S2. Taletrectinib Efficacy in ROS1^{+} NSCLC

Responses	TKI-Naïve (n=25) (%)	TKI-Pretreated (n=21) (%)
$cORR_{IRC}$, % (95% CI)[a]	92.0 (74.0, 99.0)	57.1 (34.0, 78.2)
DCR, % (95% CI)[a]	96.0 (79.7, 99.9)	85.7 (63.7, 97.0)
Median TTR, months (95% CI)	1.3 (1.3, 1.4)	1.4 (1.2, 1.6)

[a]Confidence intervals were calculated using the Clopper–Pearson method.

Table S3. Patients with TEAEs (≥15%): Taletrectinib (Pooled Safety Population; n=355)[a]

TEAE, n (%)	Patients (N=355)	
	Any Grade n (%)	Grade ≥3 n (%)
Overall	353 (99.4)	179 (50.4)
AST increased	229 (64.5)	23 (6.5)
ALT increased	212 (59.7)	32 (9.0)
Diarrhea	208 (58.6)	11 (3.1)
Nausea	157 (44.2)	5 (1.4)
Vomiting	155 (43.7)	4 (1.1)
Anemia	110 (31.0)	10 (2.8)
Dizziness	68 (19.2)	1 (0.3)
Constipation	59 (16.6)	1 (0.3)
Decreased appetite	57 (16.1)	1 (0.3)
Blood creatinine increased	57 (16.1)	0
Dysgeusia	56 (15.8)	0
Neutrophil count decreased	56 (15.8)	17 (4.8)
WBC count decreased	55 (15.5)	5 (1.4)
Blood bilirubin increased	54 (15.2)	4 (1.1)

- TEAEs led to dose reduction in 22% of patients
- TEAEs led to dose discontinuation in 5.4% of patients

[a]Pooled analysis of safety data from 6 studies in patients treated with at least one dose of taletrectinib.

AnHeart Therapeutics

Fig S1: Patient Demographics and Baseline Characteristics[a] (Safety Population)

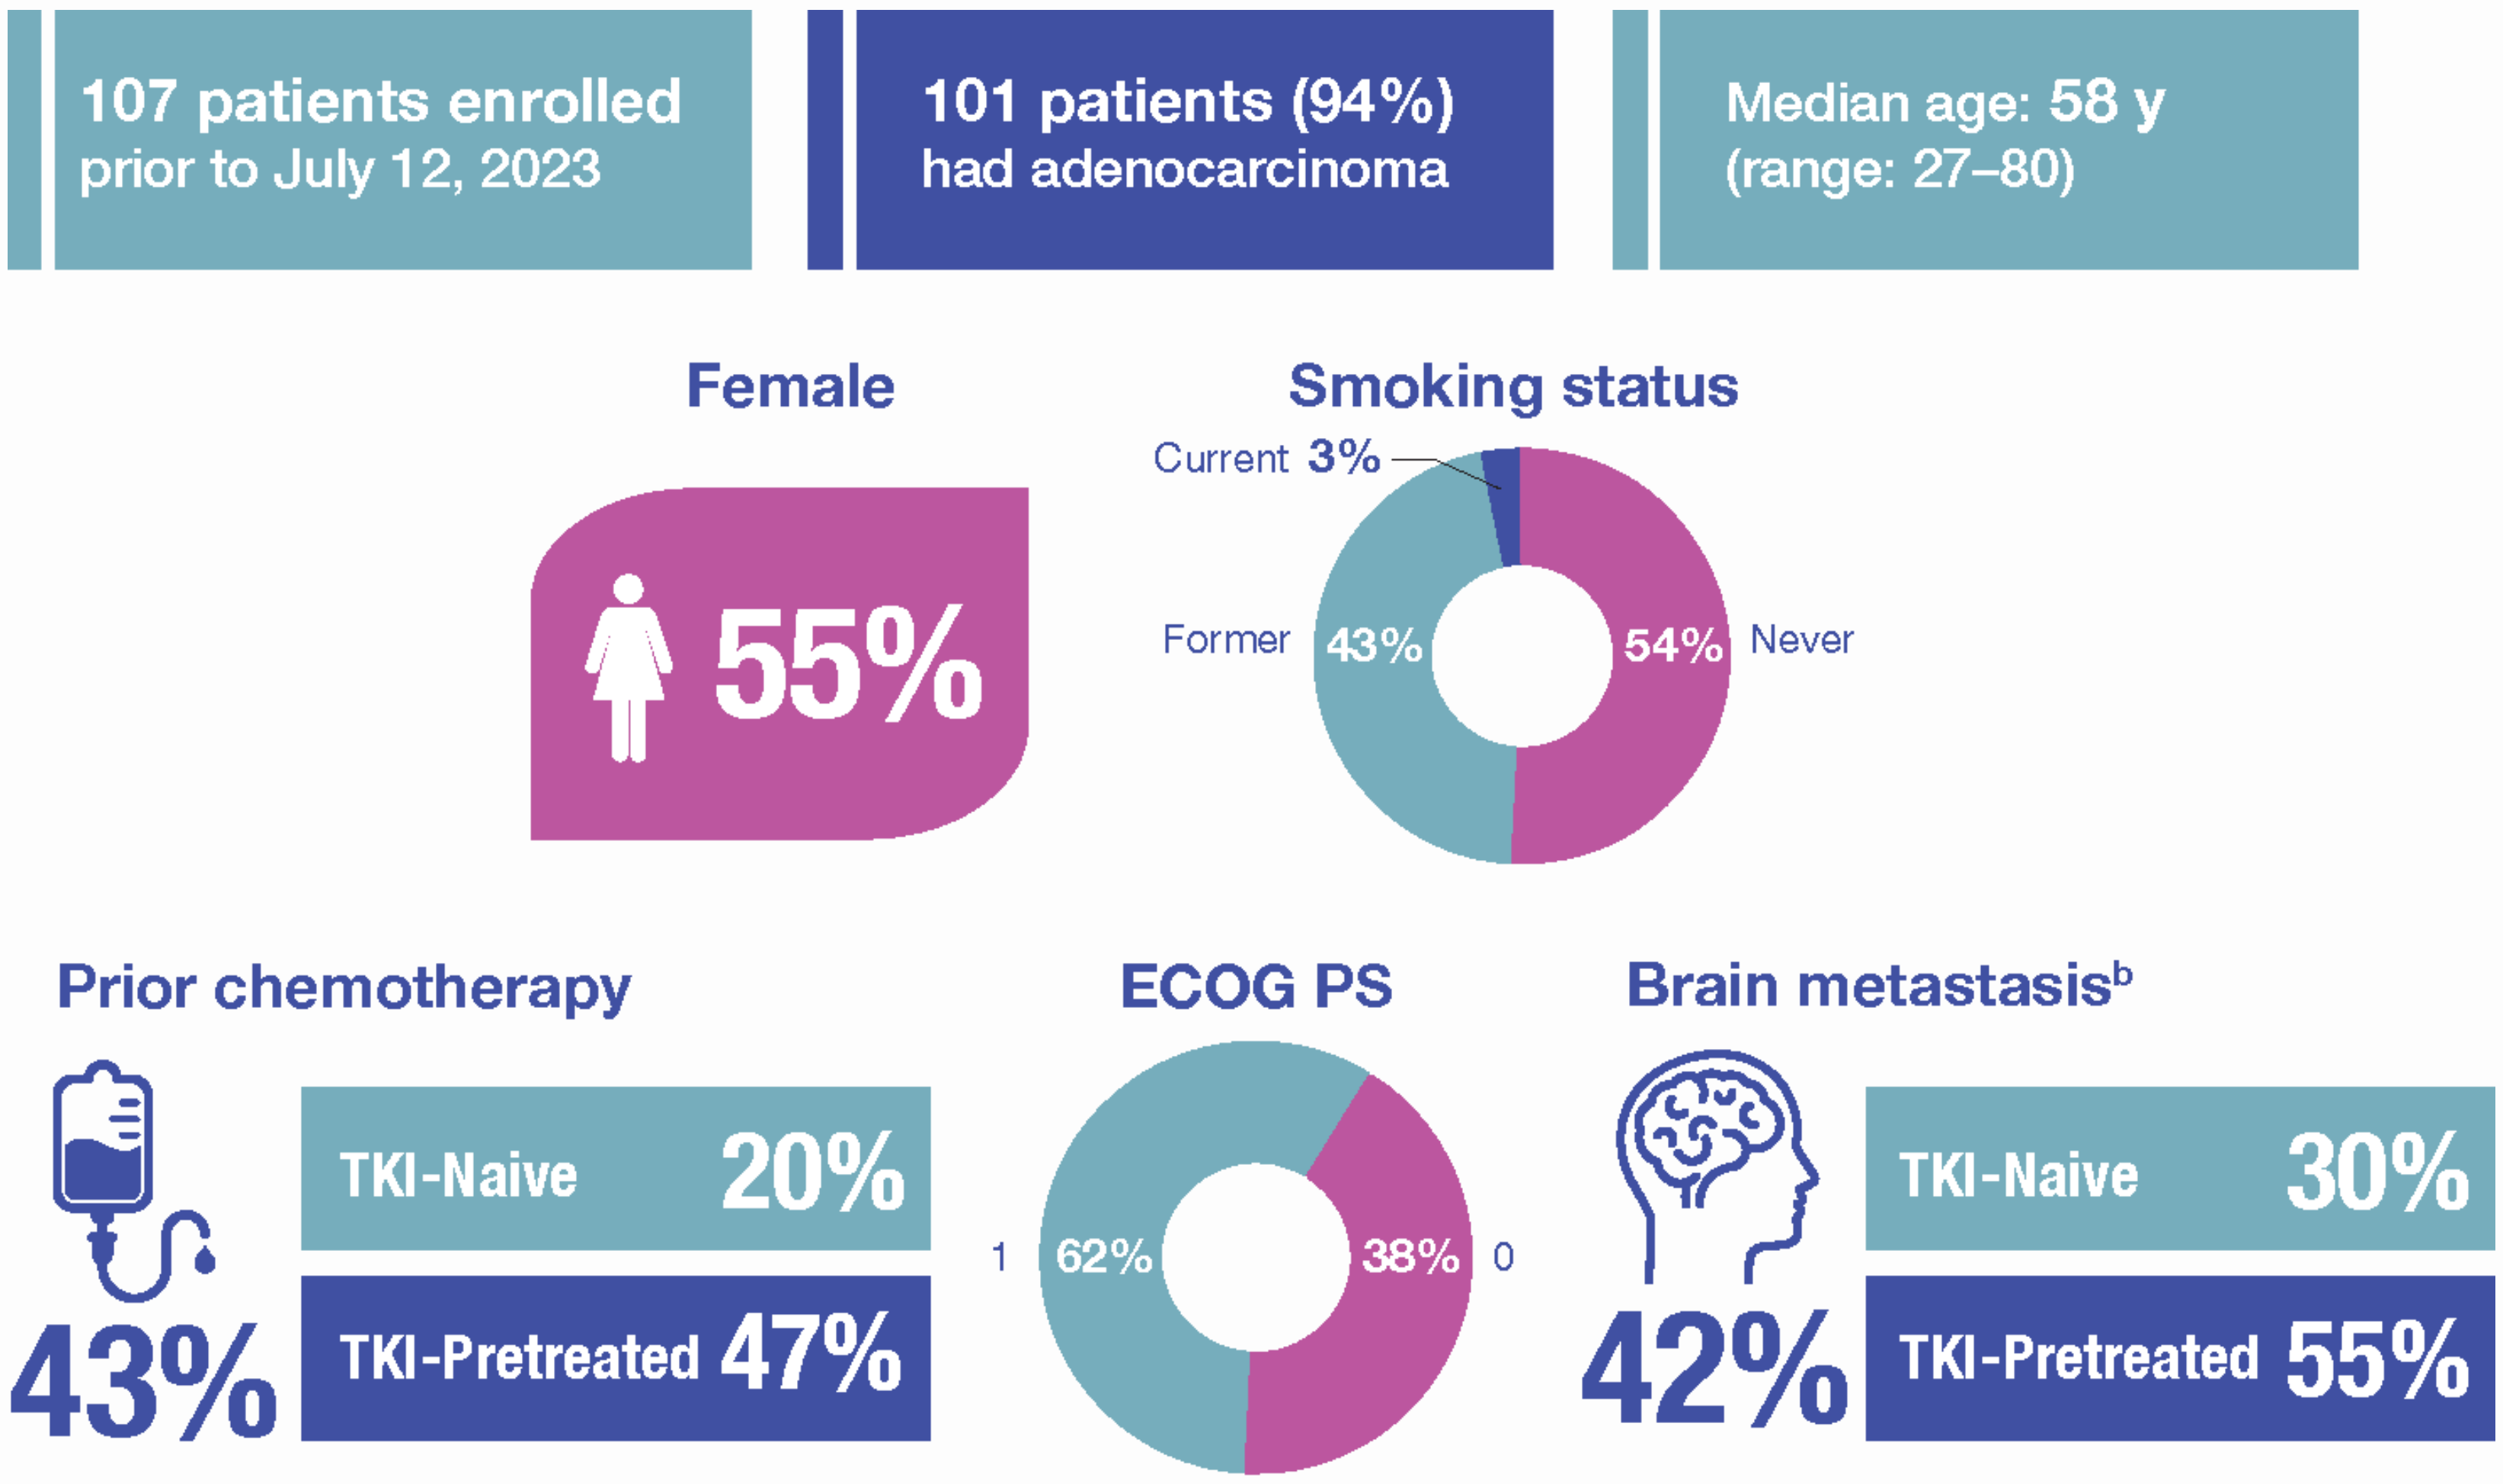
107 patients enrolled
prior to July 12, 2023
101 patients (94%)
had adenocarcinoma
Median age: 58 y
(range: 27–80)
Female
55%
Smoking status
Current 3%
Former 43%
54% Never
Prior chemotherapy
43%
TKI-Naive 20%
TKI-Pretreated 47%
ECOG PS
1 62%
38% 0
Brain metastasis[b]
42%
TKI-Naive 30%
TKI-Pretreated 55%

[a]The safety population included all patients who received ≥1 dose of taletrectinib in cohort 1 (n=44), cohort 2 (n=38), cohort 3 (n=22), and cohort 4 (n=3).
[b]Brain metastasis status by IRC (RANO-BM).

Figure S2. Patient Responses Over Time

TKI-Naive

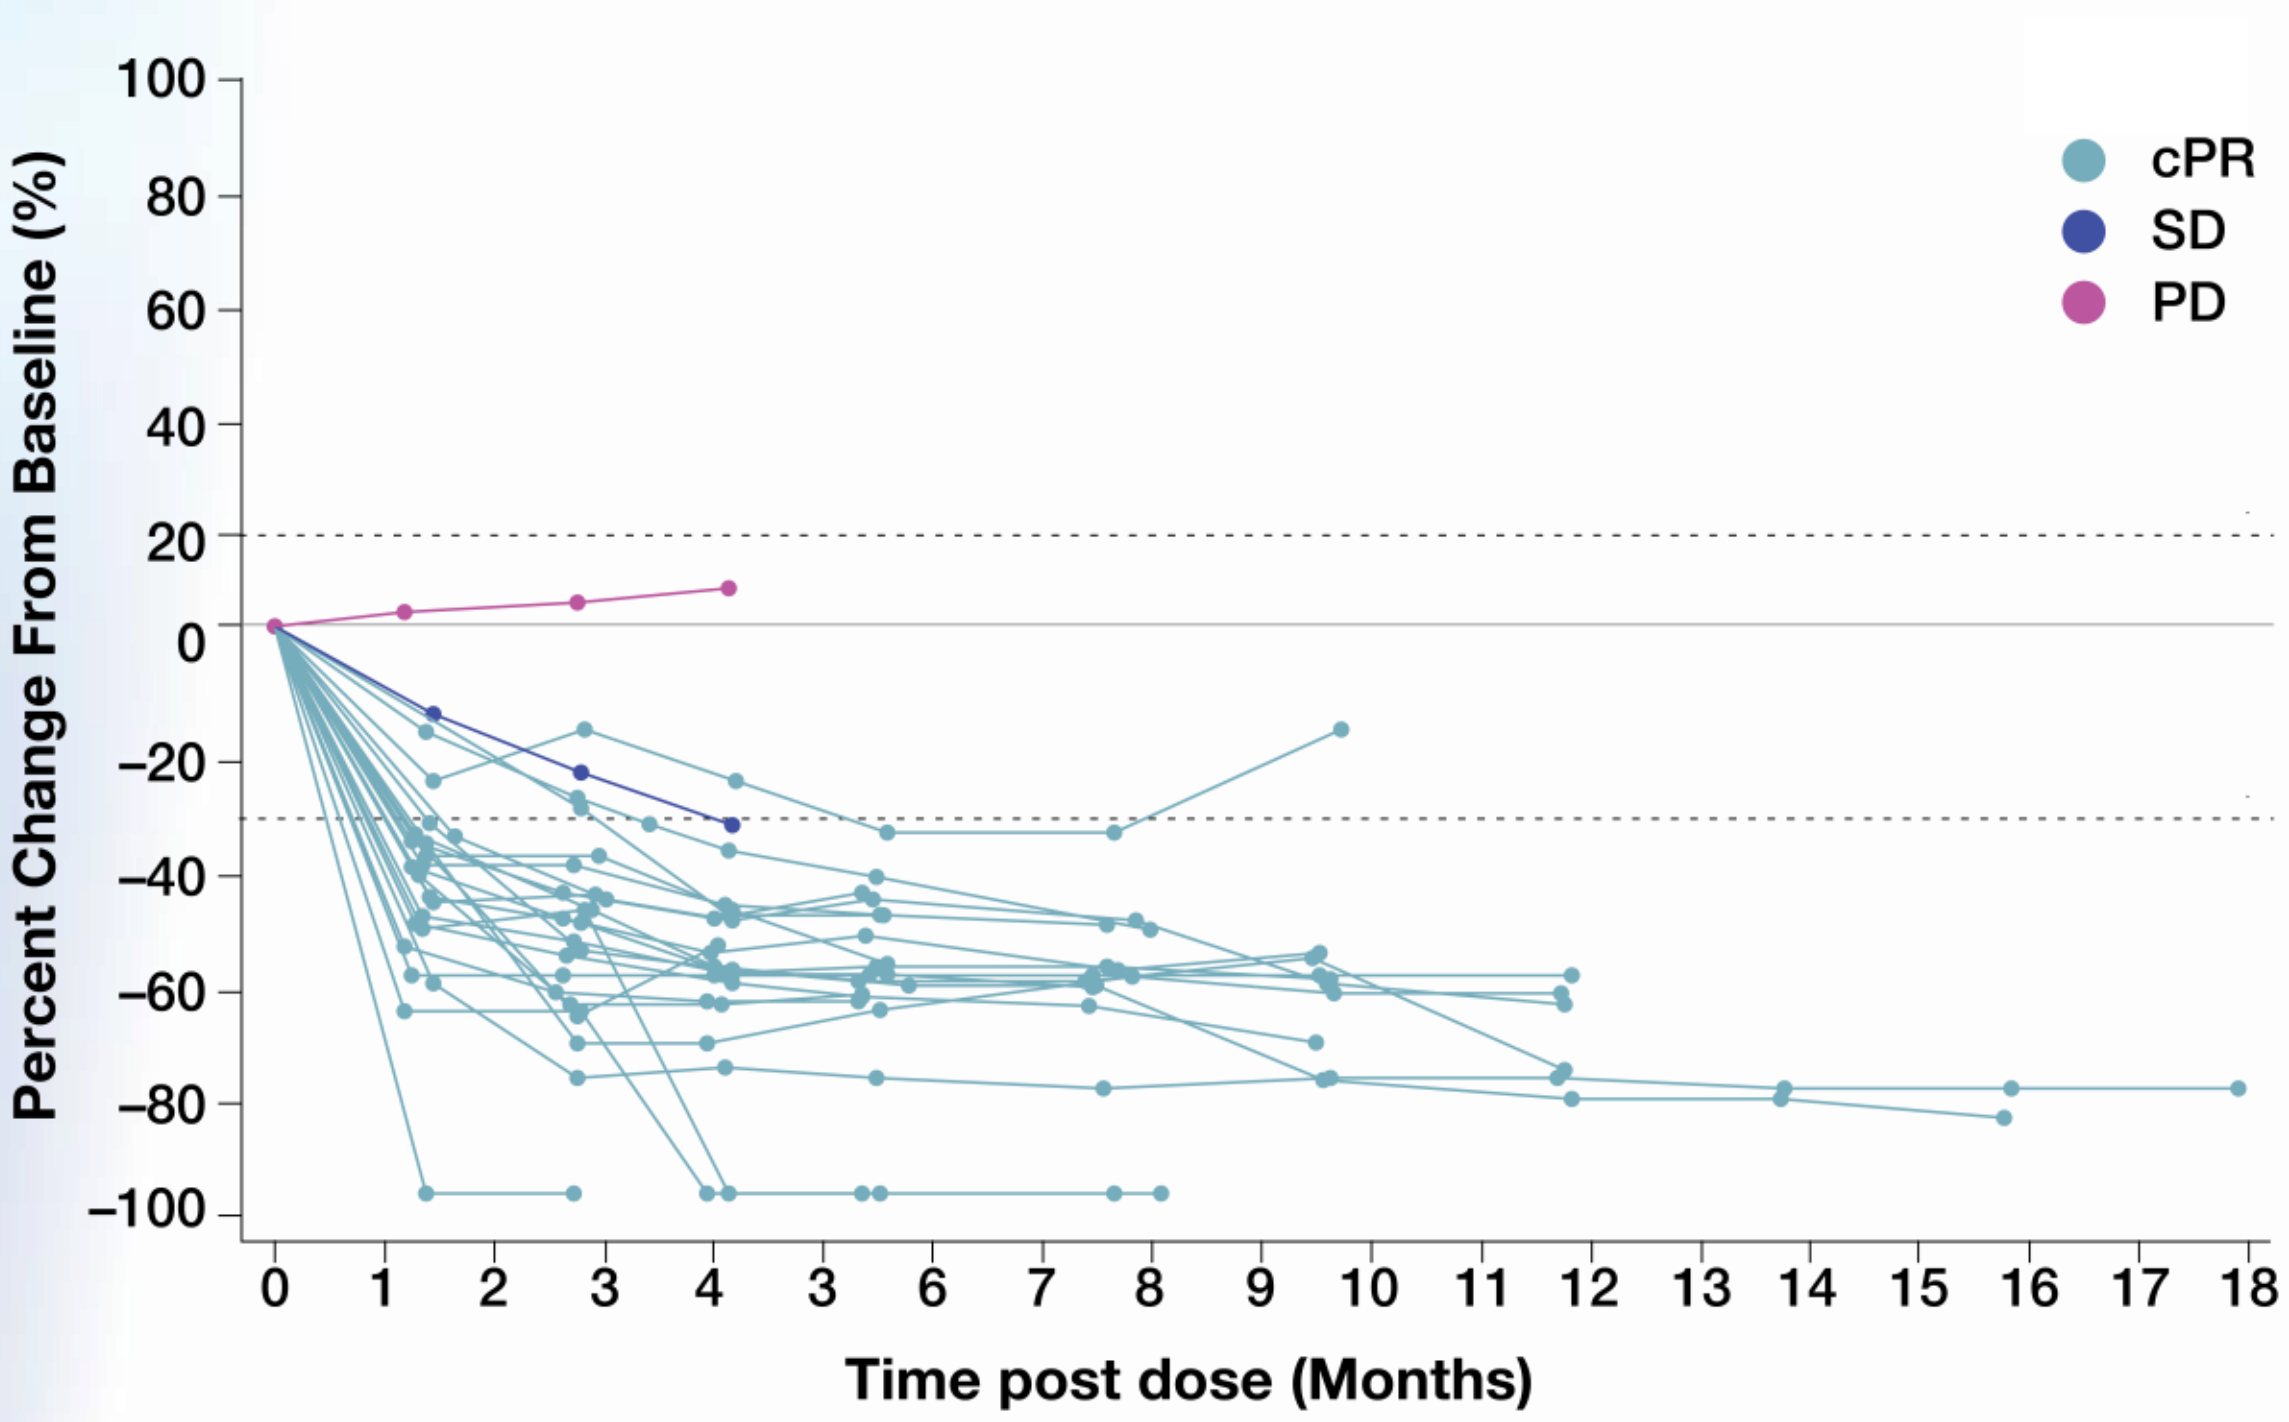

Percent Change From Baseline (%)
100
80
60
40
20
0
−20
−40
−60
−80
−100
cPR
SD
PD
0 1 2 3 4 3 6 7 8 9 10 11 12 13 14 15 16 17 18
Time post dose (Months)

TKI-Pretreated

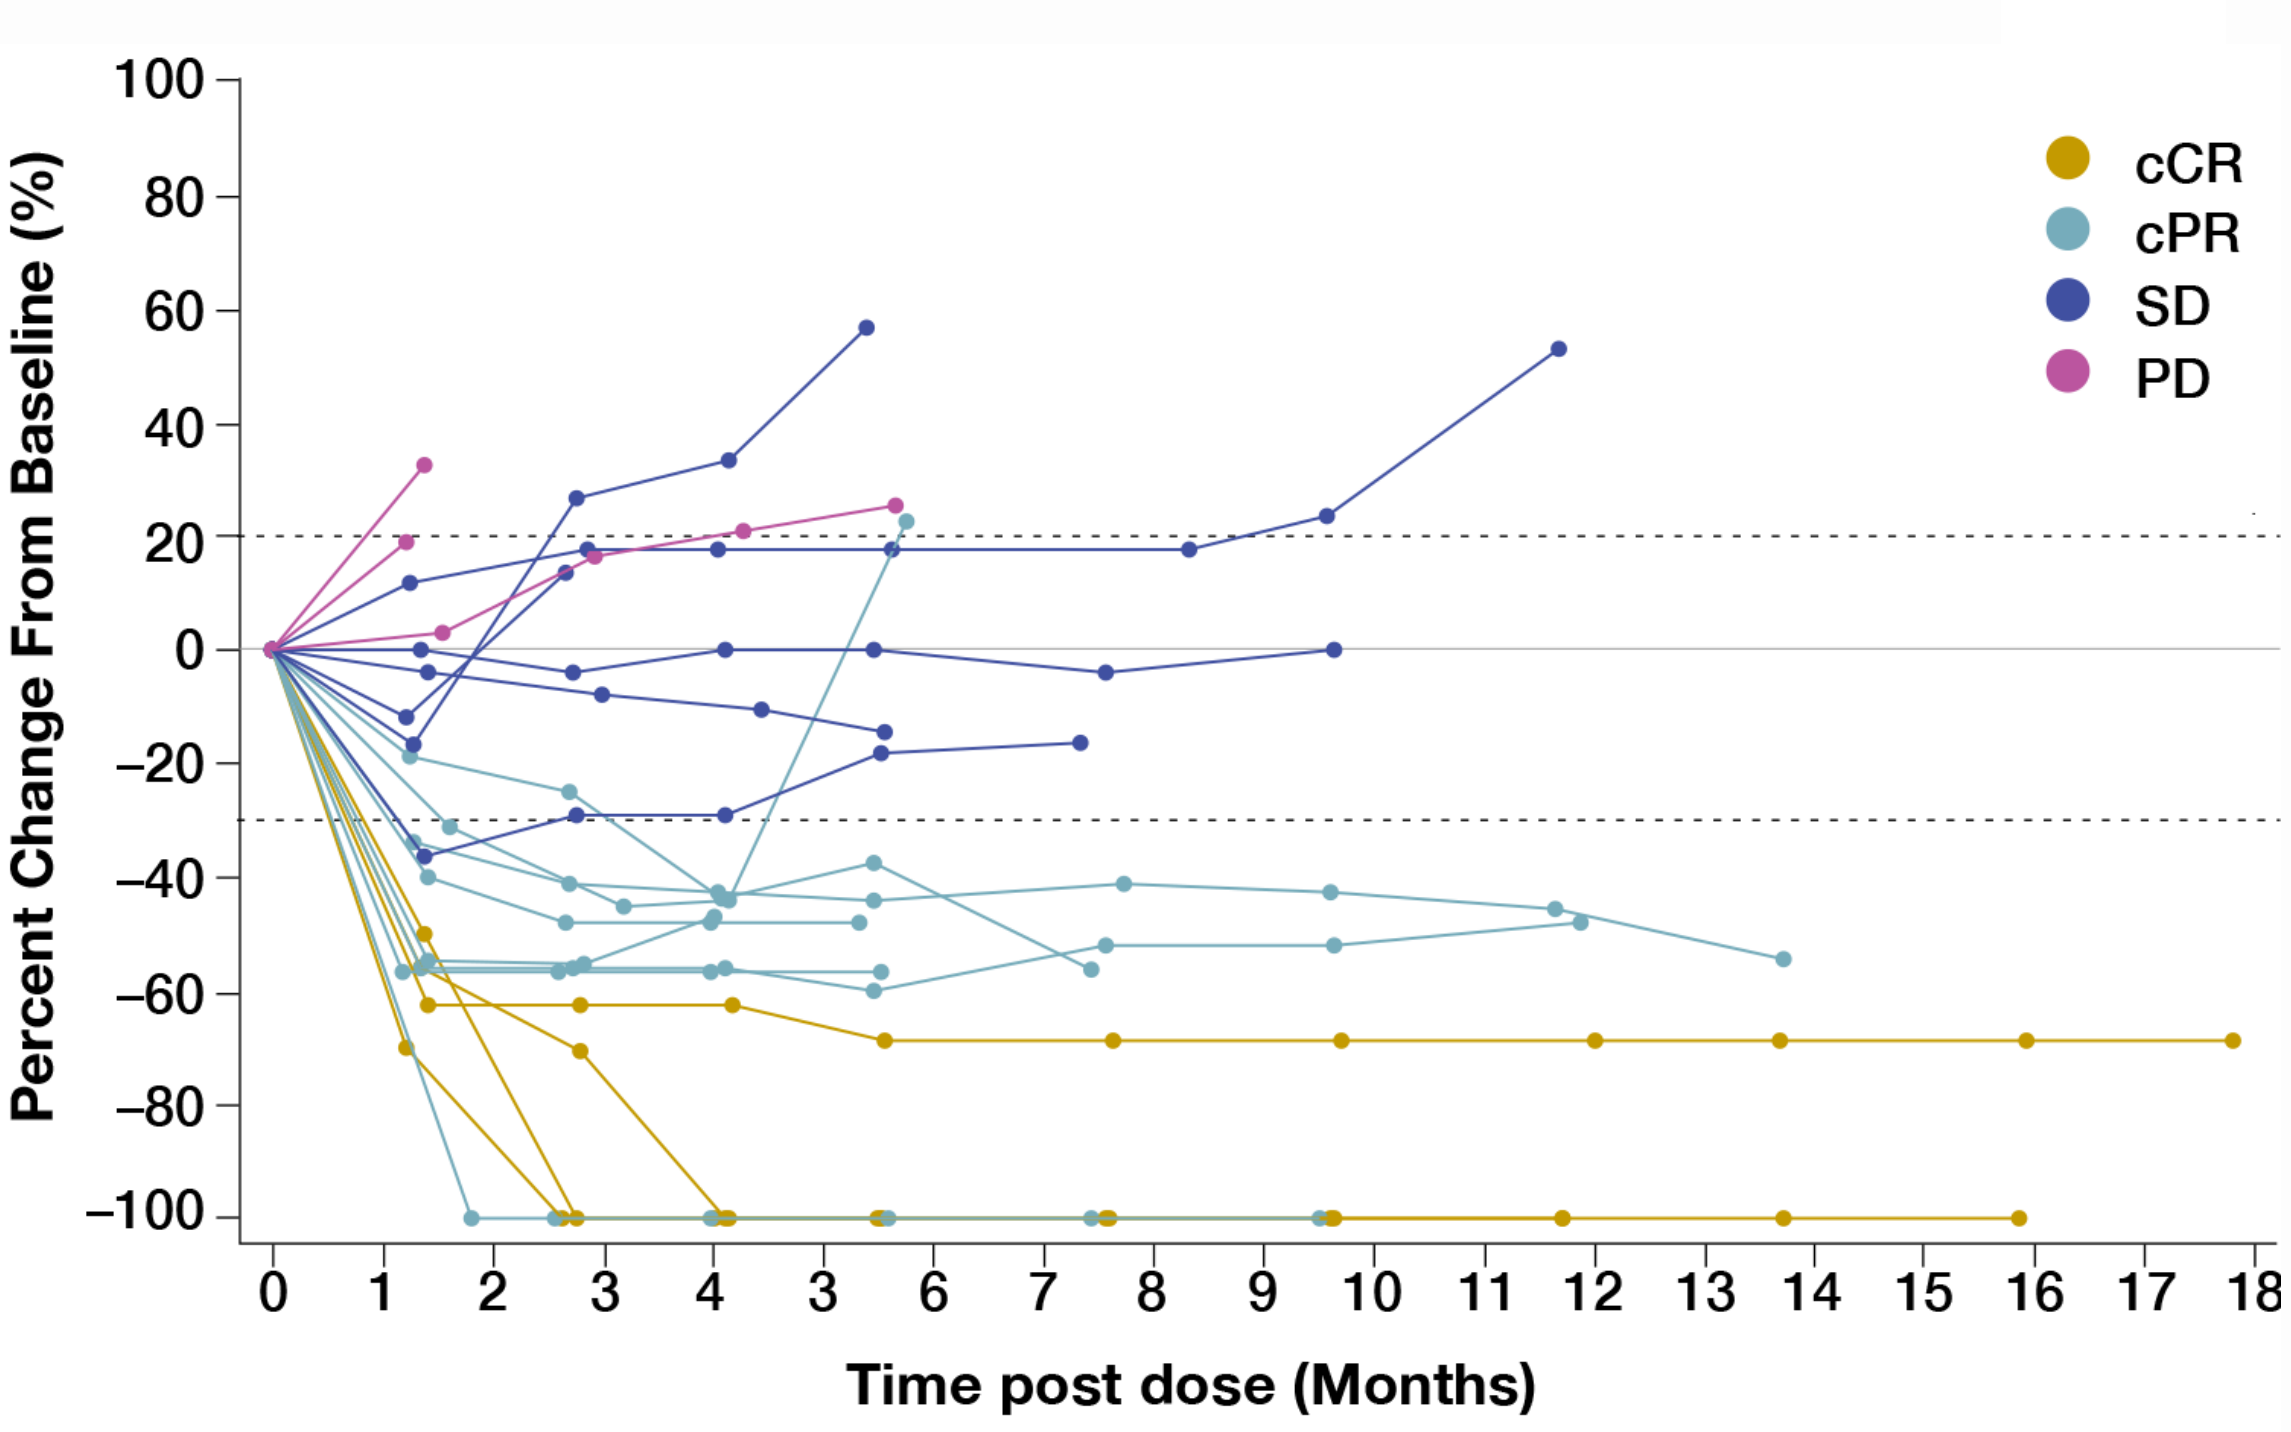

Percent Change From Baseline (%)
100
80
60
40
20
0
−20
−40
−60
−80
−100
cCR
cPR
SD
PD
0 1 2 3 4 3 6 7 8 9 10 11 12 13 14 15 16 17 18
Time post dose (Months)

AnHeart Therapeutics

Figure S3: Best Overall Response by Subgroup

TKI-Naive

Subgroup	Number of Responders/Total Number of Patients	ORR (95% CI)
Overall	23/25	92.0 (73.97–99.02)
Age		
<65 Years	16/16	100.0 (79.41–100.00)
≥65 Years	7/9	77.8 (39.99–97.19)
Gender		
Male	12/14	85.7 (57.19–98.22)
Female	11/11	100.0 (71.51–100.00)
ECOG PS		
0	11/11	100.0 (71.51–100.00)
1	12/14	85.7 (57.19–98.22)
Brain Metastasis at Baseline by IRC (RANO-BM)		
Yes	8/8	100.0 (63.06–100.00)
No	15/17	88.2 (63.56–98.54)
Prior chemotherapy		
With prior chemotherapy	4/4	100.0 (39.76–100.00)
Without prior chemotherapy	19/21	90.5 (69.62–98.83)
Lines of Prior Anti-cancer Therapy		
0	19/21	90.5 (69.62–98.83)
1-2	3/3	100.0 (29.24–100.00)
≥3	1/1	100.0 (2.50–100.00)
Region		
Western	5/6	83.3 (35.88–99.58)
Asian	18/19	94.7 (73.97–99.87)
Smoking Status		
Smoker	12/14	85.7 (57.19–98.22)
Non-Smoker	11/11	100.0 (71.51–100.00)

ORR (95% CI)

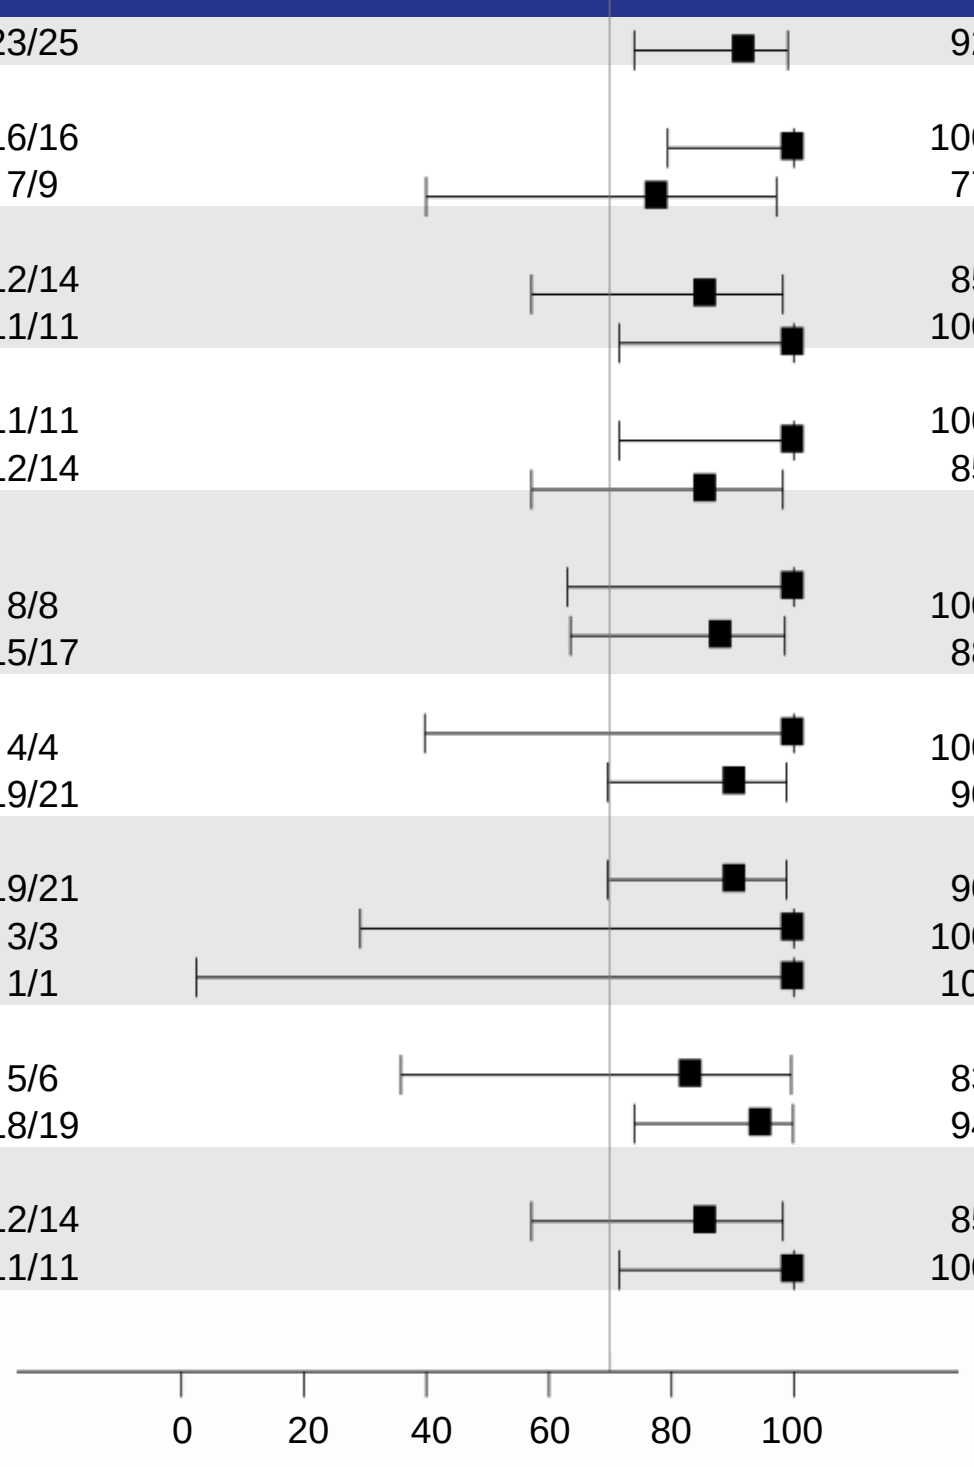
0
20
40
60
80
100

TKI-Pretreated

Subgroup	Number of Responders/Total Number of Patients	ORR (95% CI)
Overall	12/21	57.1 (34.02–78.18)
Age		
<65 Years	8/16	50.0 (24.65–75.35)
≥65 Years	4/5	80.0 (28.36–99.49)
Gender		
Male	7/11	63.6 (30.79–89.07)
Female	5/10	50.0 (18.71–81.29)
ECOG PS		
0	5/6	83.3 (35.88–99.58)
1	7/15	46.7 (21.27–73.41)
Brain Metastasis at Baseline by IRC (RANO-BM)		
Yes	5/11	45.5 (16.75–76.62)
No	7/10	70.0 (34.75–93.33)
Prior chemotherapy		
With prior chemotherapy	8/11	72.7 (39.03–93.98)
Without prior chemotherapy	4/10	40.0 (12.16–73.76)
Lines of Prior Anti-cancer Therapy		
0	0	
1-2	7/16	43.8 (19.75–70.12)
≥3	5/5	100.0 (47.82–100.00)
Region		
Western	4/6	66.7 (22.28–95.67)
Asian	8/15	53.3 (26.59–78.73)
Smoking Status		
Smoker	5/9	55.6 (21.20–86.30)
Non-Smoker	7/12	58.3 (27.67–84.83)

ORR (95% CI)

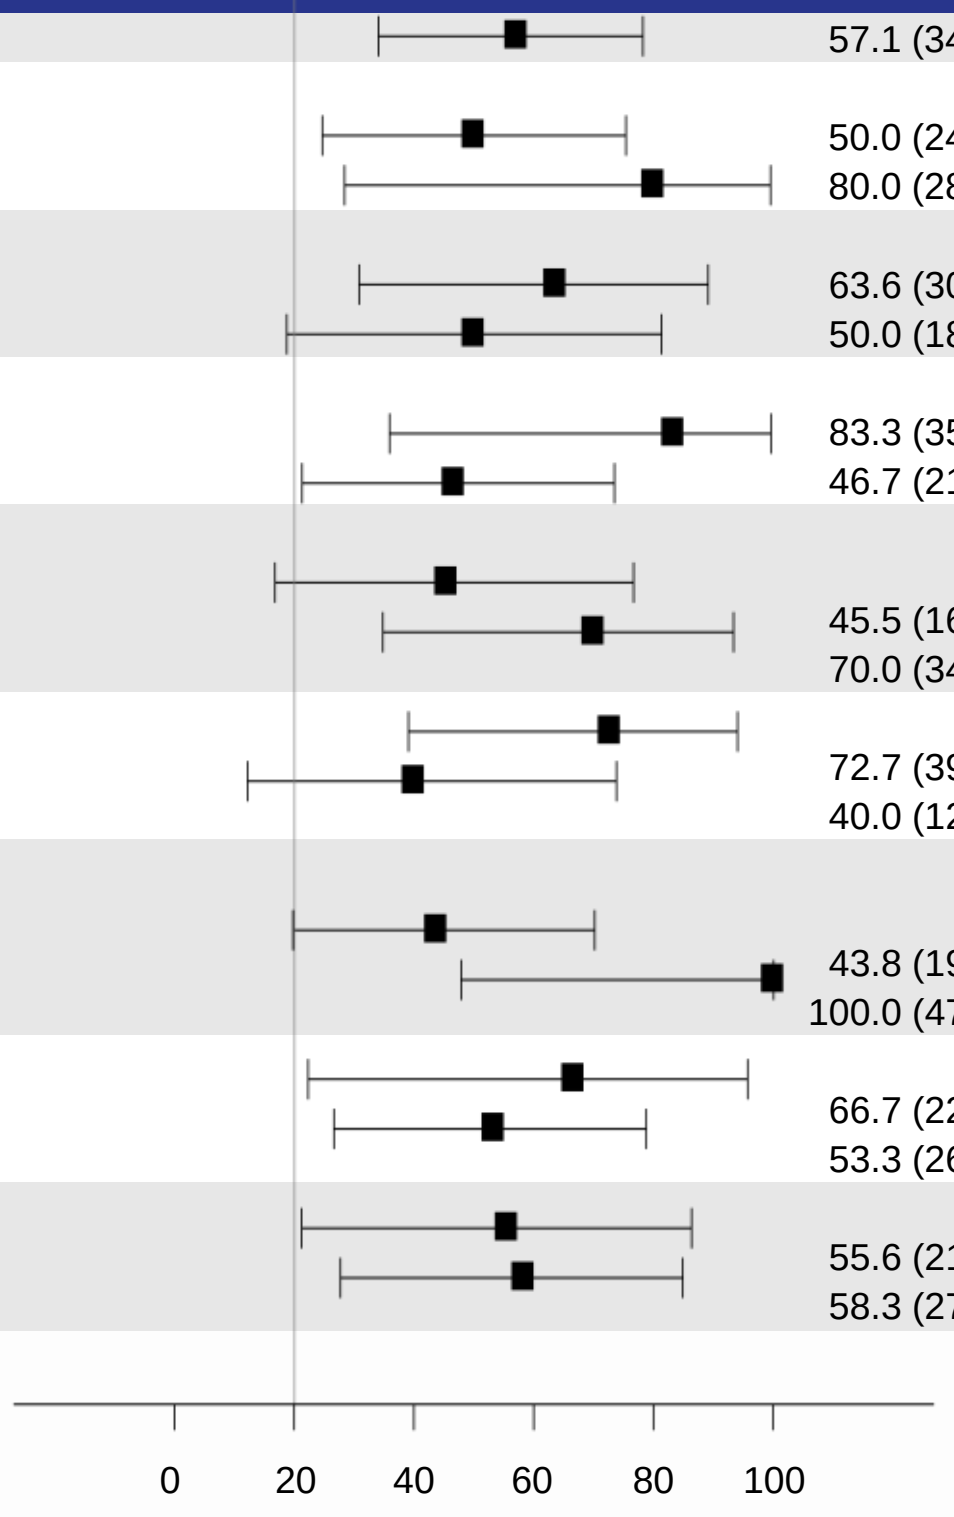
0
20
40
60
80
100

AnHeart Therapeutics